Application of a digital monitoring platform to track severity and progression in Huntington's disease

Cedric Simillion,[1] Florian Lipsmeier,[1] Atieh Bamdadian,[1] Anne Smith,[2] Scott A. Schobel,[3] Rosanna Tortelli,[4] Filipe B. Rodrigues,[4] Lauren M. Byrne,[4] Edward J. Wild,[4] Michael Lindemann[1]

1. Roche Pharma Research and Early Development, pRED Informatics, F. Hoffmann-La Roche Ltd, Basel, Switzerland; 2. Ionis Pharmaceuticals Inc, Carlsbad, CA, USA; 3. F. Hoffmann-La Roche Ltd, Basel, Switzerland; 4. Huntington's Disease Centre, UCL Queen Square Institute of Neurology, University College London, London, UK.

Introduction

- Huntington's disease is a rare, hereditary, progressive brain disease that causes problems with a person's ability to think, behave and move.[1,2] Progressive means it gets worse over time.
- Signs and symptoms of Huntington's are usually assessed during visits to the doctor, which may not happen often enough to continuously track how the disease evolves over time.
- Advances in wearable sensors and communication technologies have enabled monitoring of a patient's symptoms from their own home, with the use of a digital monitoring platform consisting of a smartwatch and a smartphone.[3–5]

Conclusions

- The Roche Huntington's disease digital monitoring platform allows for symptoms to be captured at home, in between scheduled visits to the doctor, and has the potential to serve as a tool that would allow doctors to monitor the evolution of a patient's disease over time, remotely and more frequently.[3–5]
- Digital tests have been inspired by disease understanding and existing tests performed at the doctor's clinic, and may help the patient and clinician to more effectively manage this disease and improve quality of life.
- The majority of patients in the open-label extension study had a positive experience with the digital monitoring platform and regularly completed most of the digital tests.

The Roche Huntington's disease digital monitoring platform is being used across the tominersen global Clinical Development Programme

- Tominersen is a medication currently in clinical development and not yet approved for Huntington's.
- It is designed to target the underlying cause of the disease and has the potential to slow or stop the progressive nature of the disease.
- Roche has a comprehensive clinical development programme investigating the dosing, safety and potential clinical benefits of tominersen in people with Huntington's.

The digital monitoring platform uses smartwatch- and smartphone-based technology to monitor the evolution of Huntington's symptoms across multiple studies in tominersen's Clinical Development Programme

Open-label extension (NCT03342053)* (46 patients)
- **Objective:** Assess safety and the body's responses to tominersen in patients with early movement symptoms over 15 months

Completed

HD Natural History Study (NCT03664804) (95 patients)
- **Objective:** Collect data on disease in individuals not receiving tominersen in patients with early movement symptoms over 15 months

GENERATION HD1 (NCT03761849) (791 patients)
- **Objective:** Investigate safety and effectiveness of tominersen in patients with movement symptoms over 25 months (plus follow-up)

GEN-EXTEND (NCT03842969)
Objective: Continue to assess safety and effectiveness of tominersen once patients have completed other studies

A study run by UCL is also using the digital monitoring platform to measure symptoms in different groups of people

Digital-HD (120 participants)
- **Objective:** Investigate the suitability of the digital monitoring platform in healthy individuals and individuals with Huntington's

* The open-label extension study was an extension of the original Phase I/IIa study of tominersen which included the same 46 patients.

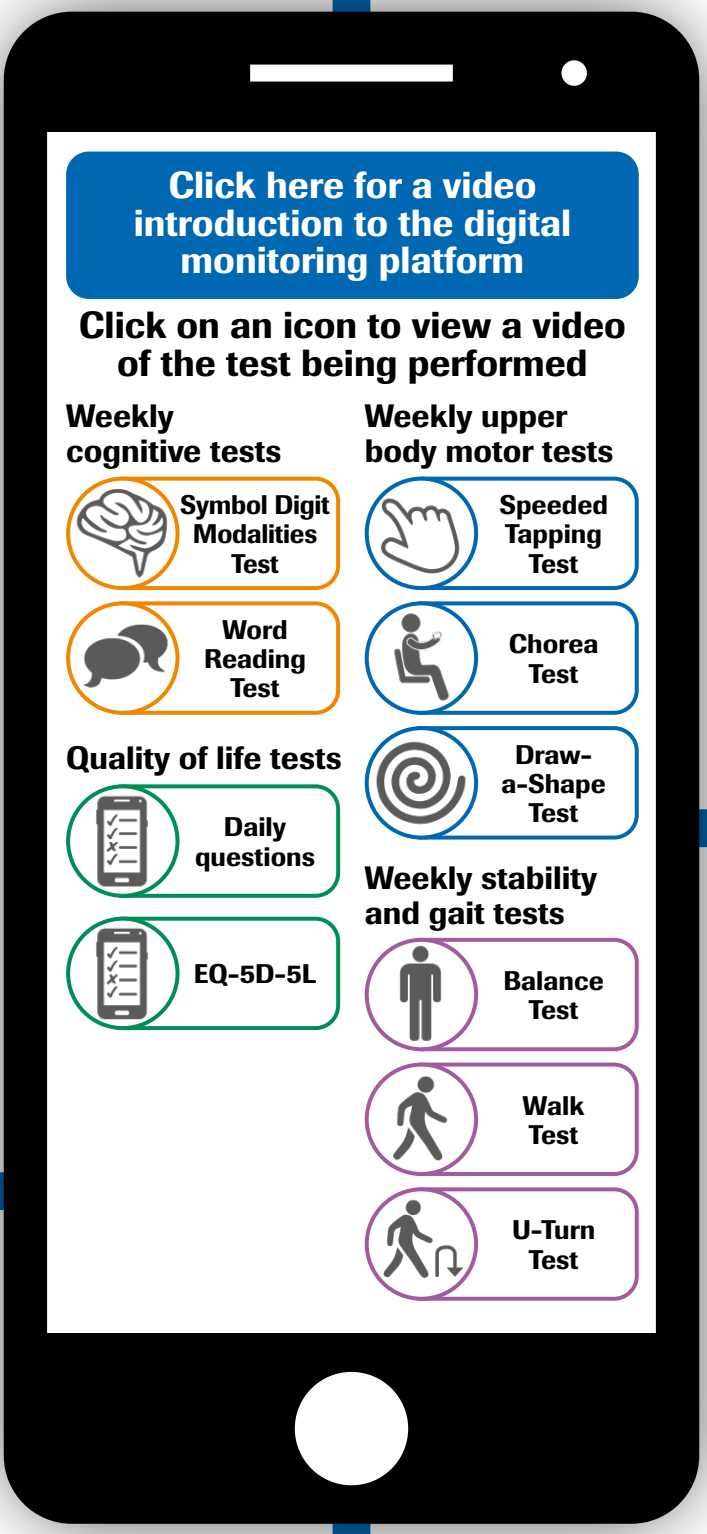
Click here for a video introduction to the digital monitoring platform
Click on an icon to view a video of the test being performed
Weekly cognitive tests
Symbol Digit Modalities Test
Word Reading Test
Quality of life tests
Daily questions
EQ-5D-5L
Weekly upper body motor tests
Speeded Tapping Test
Chorea Test
Draw-a-Shape Test
Weekly stability and gait tests
Balance Test
Walk Test
U-Turn Test

Patient experience with the digital platform during the open-label extension study was positive

72% of patients answered the satisfaction questionnaire

83% of patients were positive or neutral towards the digital monitoring platform

Taking all things into account, how satisfied or dissatisfied are you with the Huntington's study phone, watch and monitoring app?

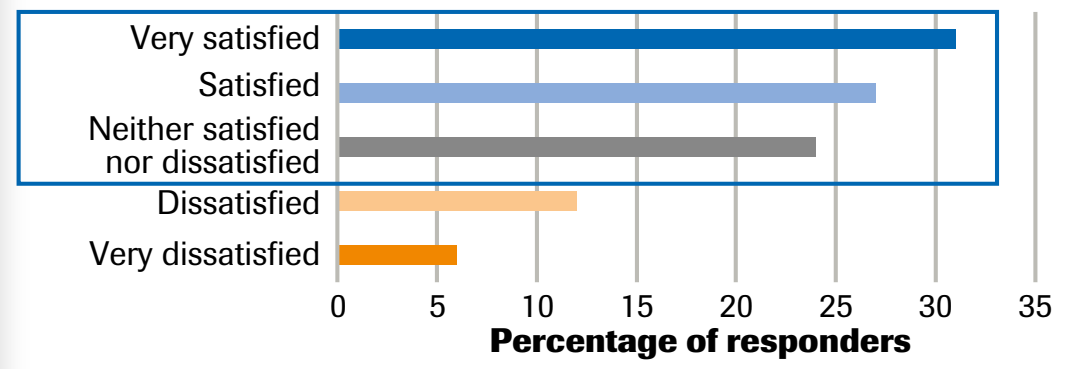
Very satisfied
Satisfied
Neither satisfied nor dissatisfied
Dissatisfied
Very dissatisfied
0
5
10
15
20
25
30
35
Percentage of responders

Participants in the open-label extension study performed daily tests on more than 5 days per week, suggesting patients were willing to use the digital monitoring platform on a regular basis

Digital upper body tests showed a difference between individuals with premanifest Huntington's and healthy individuals in the Digital-HD study[7]

- Individuals with premanifest Huntington's performed less consistently in tests involving hand and finger movements (Speeded Tapping Test and Draw-a-Shape Test) compared with healthy individuals.
- The differences in hand and finger movements identified by digital tests may allow doctors to identify subtle movement problems which can't be detected at regular visits to the clinic and which may represent very early manifestations of the disease.

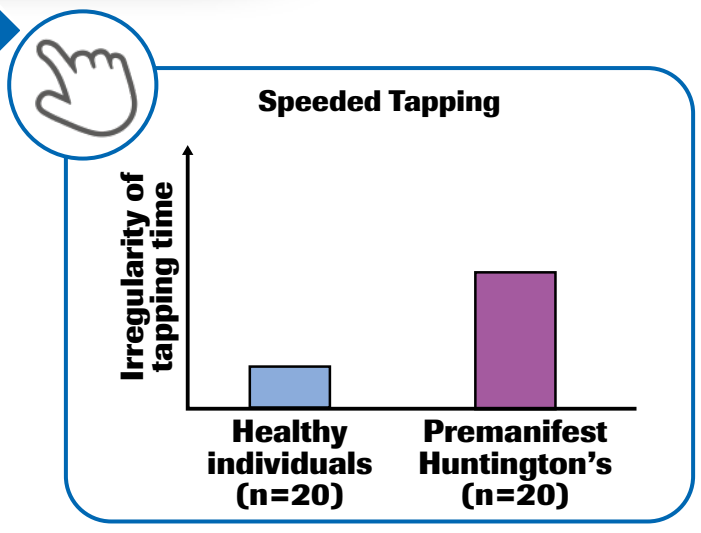
Speeded Tapping
Irregularity of tapping time
Healthy individuals (n=20)
Premanifest Huntington's (n=20)

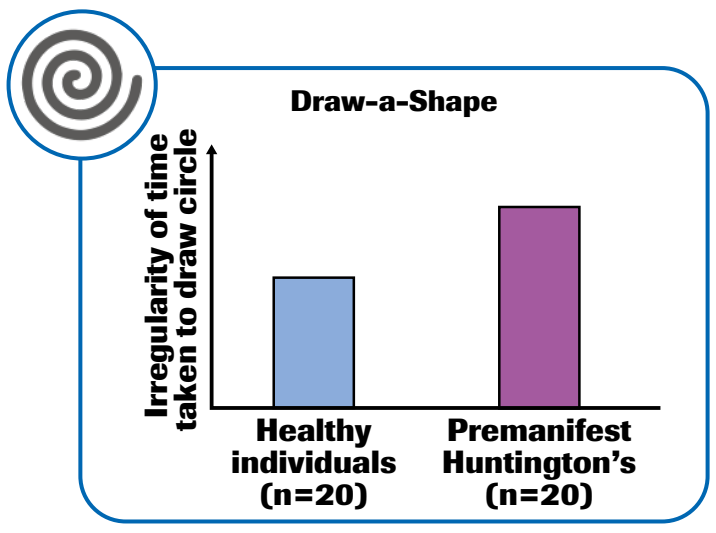
Draw-a-Shape
Irregularity of time taken to draw circle
Healthy individuals (n=20)
Premanifest Huntington's (n=20)

* Figures adapted from Tortelli R, et al. *Neurotherapeutics*. 2019; 16:1350–1390:S74.

Performance in digital tests worsens as Huntington's progresses

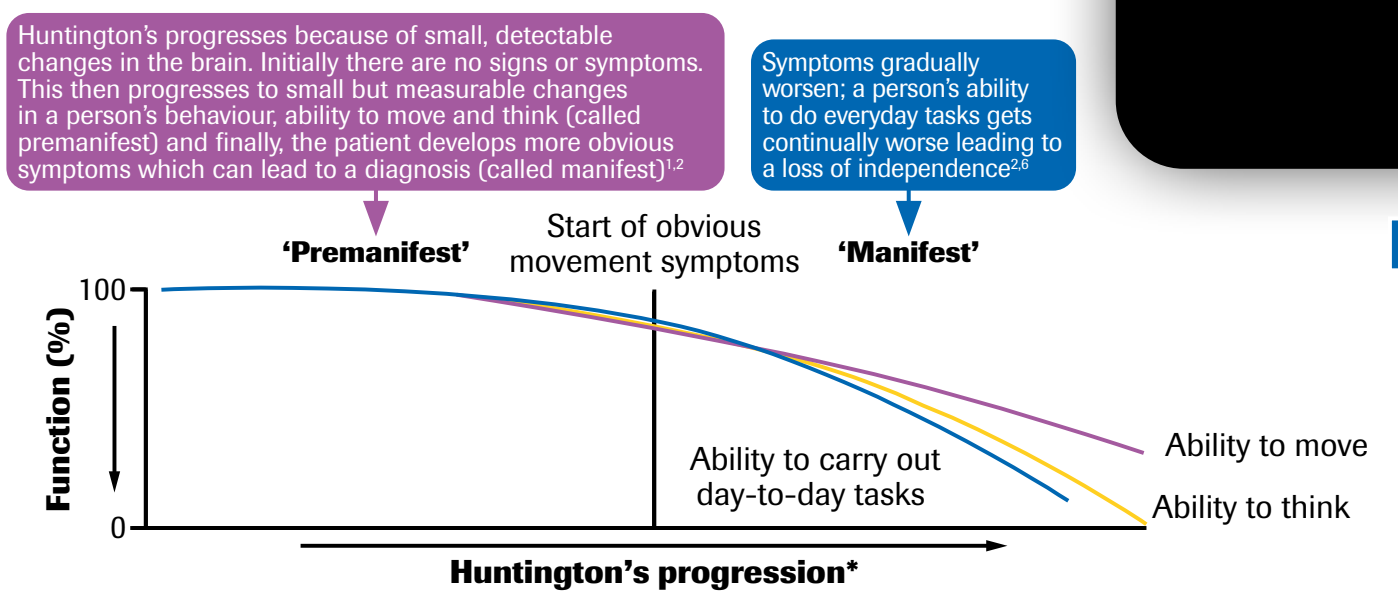
Huntington's progresses because of small, detectable changes in the brain. Initially there are no signs or symptoms. This then progresses to small but measurable changes in a person's behaviour, ability to move and think (called premanifest) and finally, the patient develops more obvious symptoms which can lead to a diagnosis (called manifest)1,2
Symptoms gradually worsen; a person's ability to do everyday tasks gets continually worse leading to a loss of independence2,6
'Premanifest'
Start of obvious movement symptoms
'Manifest'
Function (%)
100
0
Ability to move
Ability to think
Ability to carry out day-to-day tasks
Huntington's progression*

- The results of the Digital-HD study have shown that individuals with manifest Huntington's had reduced performance in most digital tests compared to those with premanifest Huntington's and healthy individuals.[7]
- These results suggest that the digital monitoring platform can effectively monitor the evolution of Huntington's symptoms, which has the potential to help patients and clinicians to more effectively manage this disease and improve quality of life.

* Figure adapted from Ross CA, et al. *Nat Rev Neurol*. 2014; 10:204–216 and Walker FO. *Lancet*. 2007; 369:218–228.

Acknowledgements

We thank all the patients who participate in our studies and their families. Ionis Pharmaceuticals discovered tominersen and is partnered with F. Hoffmann La-Roche for its development. The OLE study was initially sponsored by Ionis Pharmaceuticals, and transferred to F. Hoffmann-La Roche Ltd. The HD NHS and GENERATION HD1 studies are funded by F. Hoffmann-La Roche Ltd. The Digital-HD study is sponsored by the University College London (UCL), supported by a research grant to UCL from F. Hoffmann-La Roche Ltd. The authors thank Sarah Child, of Meditech Media UK for providing editorial support for this poster, which was funded by F. Hoffmann-La Roche Ltd in accordance with Good Publication Practice (GPP3) guidelines (http://www.ismpp.org/gpp3).

References

1. Bates GP, et al. *Nat Rev Dis Primers*. 2015; 1:15005; 2. Ross CA, et al. *Nat Rev Neurol*. 2014; 10:204–216; 3. Lipsmeier F, et al. *Mov Disord*. 2018; 33:1287–1297; 4. Lipsmeier F, et al. *J Neurol Neurosurg Psychiatry*. 2018; 89:F61; 5. Lipsmeier F, et al. *Neurology*. 2019; 92:S15; 6. Walker FO. *Lancet*. 2007; 369:218–228; 7. Tortelli R, et al. *Neurotherapeutics*. 2019; 16:1350–1390:S74.

Please scan using your QR reader application to access this poster on your mobile device. NB: there may be associated costs for downloading data. These costs may be high if you are using your smartphone abroad. Please check your mobile data tariff or contact your service provider for more details. Alternatively this can be accessed at: https://bit.ly/2X0nABK

Presented at the 10th European Conference on Rare Diseases & Orphan Products – Virtual Meeting; 15–16 May, 2020