Poster Number 173

PROJECT HERCULES: THE CHALLENGES OF ESTIMATING MULTI-STATE MODEL TRANSITIONS IN RARE DISEASES: CREATING A NATURAL HISTORY MODEL FOR DUCHENNE MUSCULAR DYSTROPHY (DMD)

Broomfield J[1*], Hill M[1], Crowther MJ[1,2], Larkindale J[3], Guglieri M[4], Godfrey J[5], Chandler F[5], Abrams KR[1], [on behalf of Project HERCULES]

[1] Department of Health Sciences, University of Leicester, Leicester, UK *Contact: jb781@le.ac.uk

Theme 5: Achieving the triple A's by 2030: Accessible, Available and Affordable Treatments for people living with a rare disease.

Introduction

About Duchenne

Duchenne Muscular Dystrophy (DMD) is a rare progressive disease that is often diagnosed in early childhood, and results in significantly reduced life expectancy. The disease almost always affects boys, and there is no current cure.

About Project HERCULES

Project HERCULES is an international multi-stakeholder collaboration led by Duchenne UK that is developing disease-level tools and evidence to support HTA and access decisions for new treatments for Duchenne Muscular Dystrophy.

About The Research

Due to the rarity of DMD, both the research literature and patient numbers are limited. This study aimed to fully characterise the natural history of DMD by synthesising data from clinical trials, digital extraction from the literature, and elicitation from stakeholders. A unified analysis was performed to represent the progression of a patient with DMD and to inform future economic decision models on potential treatments.

Figure 1

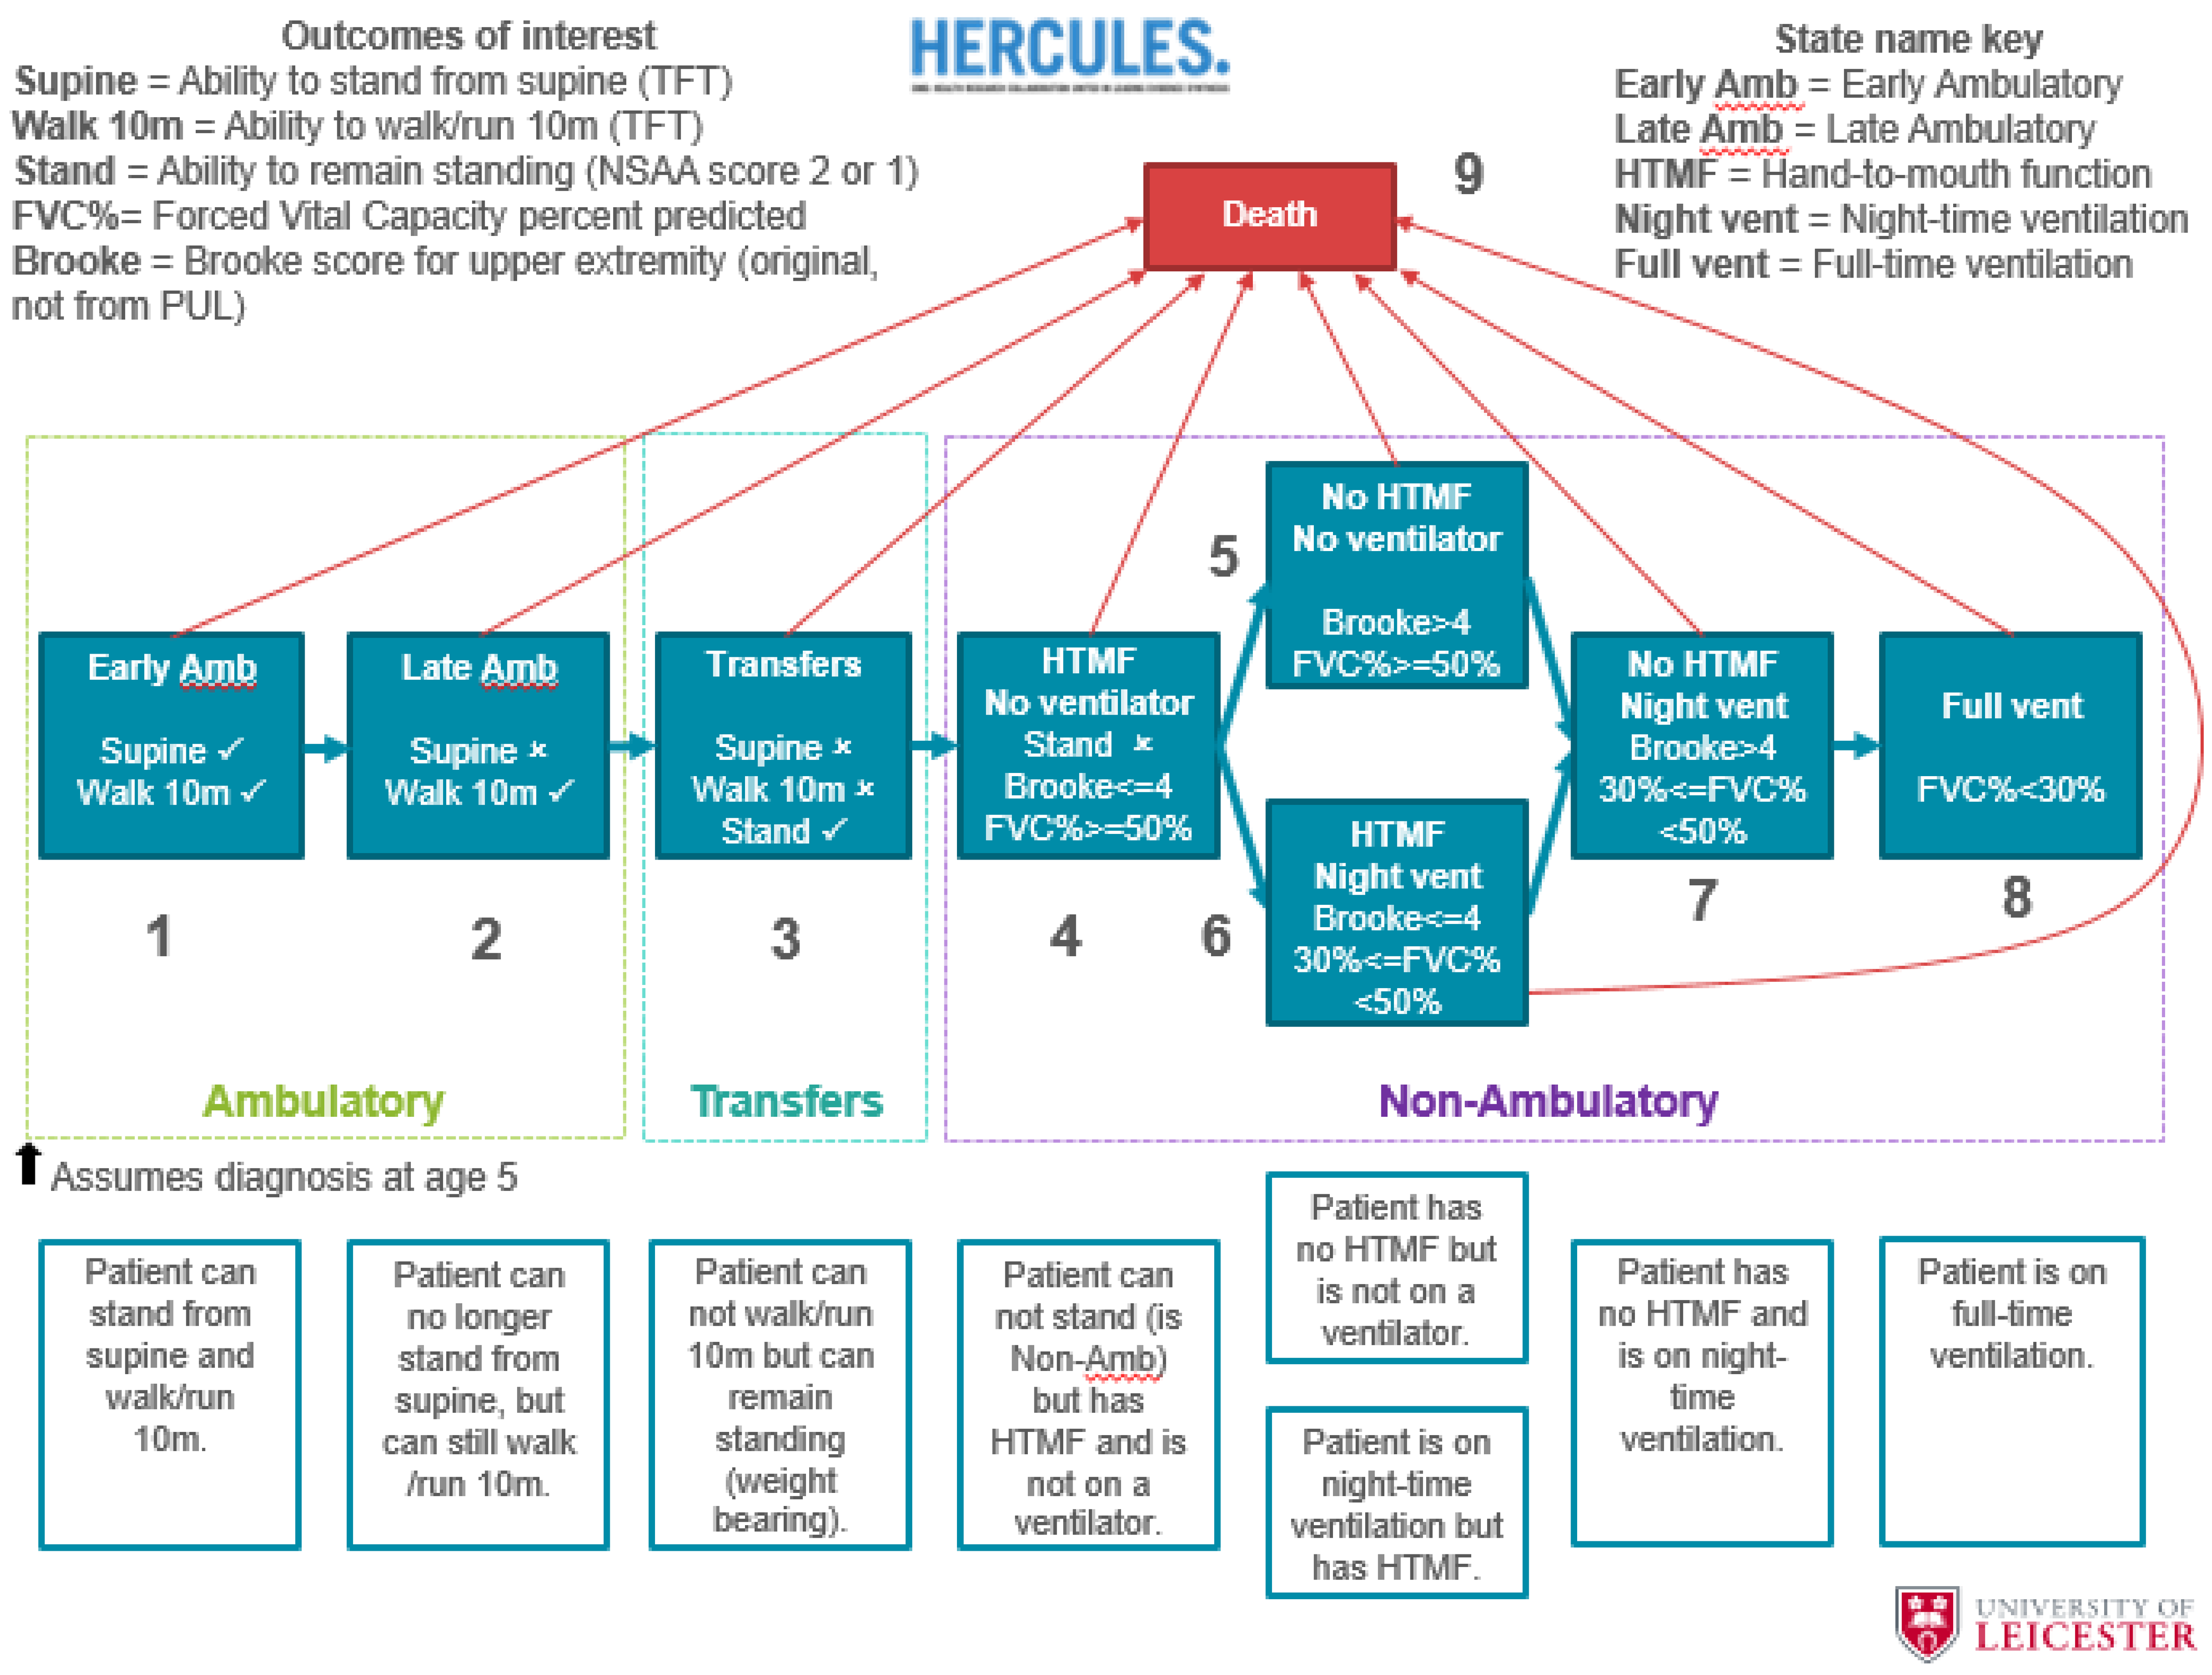
Outcomes of interest
Supine = Ability to stand from supine (TFT)
Walk 10m = Ability to walk/run 10m (TFT)
Stand = Ability to remain standing (NSAA score 2 or 1)
FVC%= Forced Vital Capacity percent predicted
Brooke = Brooke score for upper extremity (original, not from PUL)
HERCULES.
State name key
Early Amb = Early Ambulatory
Late Amb = Late Ambulatory
HTMF = Hand-to-mouth function
Night vent = Night-time ventilation
Full vent = Full-time ventilation
Death
9
Early Amb
Supine ✓
Walk 10m ✓
1
Late Amb
Supine ×
Walk 10m ✓
2
Transfers
Supine ×
Walk 10m ×
Stand ✓
3
HTMF
No ventilator
Stand ×
Brooke<=4
FVC%>=50%
4
No HTMF
No ventilator
Brooke>4
FVC%>=50%
5
HTMF
Night vent
Brooke<=4
30%<=FVC%
<50%
6
No HTMF
Night vent
Brooke>4
30%<=FVC%
<50%
7
Full vent
FVC%<30%
8
Ambulatory
Transfers
Non-Ambulatory
Assumes diagnosis at age 5
Patient can stand from supine and walk/run 10m.
Patient can no longer stand from supine, but can still walk /run 10m.
Patient can not walk/run 10m but can remain standing (weight bearing).
Patient can not stand (is Non-Amb) but has HTMF and is not on a ventilator.
Patient has no HTMF but is not on a ventilator.
Patient is on night-time ventilation but has HTMF.
Patient has no HTMF and is on night-time ventilation.
Patient is on full-time ventilation.
UNIVERSITY OF LEICESTER

Figure 1: Proposed natural history model for Duchenne Muscular Dystrophy.

Methods

Natural history model

The proposed model in Figure 1 is the result of multiple stakeholder input from clinicians, patients and carers. State definitions were made in accordance primarily with clinical beliefs, while also bearing the (limited) data availability in mind.

Data sources

- **Intermediate transitions:**

Information on transitions from states 1 to 8 were available from clinical trials (mainly from placebo arms, made accessible by the C-Path Institute[1]).

- **Mortality transitions:**

No data was available on transitions to state 9: instead, a literature review was conducted identifying appropriate Kaplan-Meier survival curves, from which survival data was extracted via Guyot's algorithm[2].

- **Transfer state (state 3) transitions:**

Clinical trials did not report necessary information; instead, these transitions were informed by simulated data based on clinical, carer and patient elicitation.

Analysis

A parametric Markov multi-state model was fitted to the combined dataset with age as the time-scale, assuming constant rates of transition between states and supplemented by repetitively estimated mortality rates. The model fit was iterated until convergence of all transition rates. This was repeated for a steroid-user population.

Figure 2

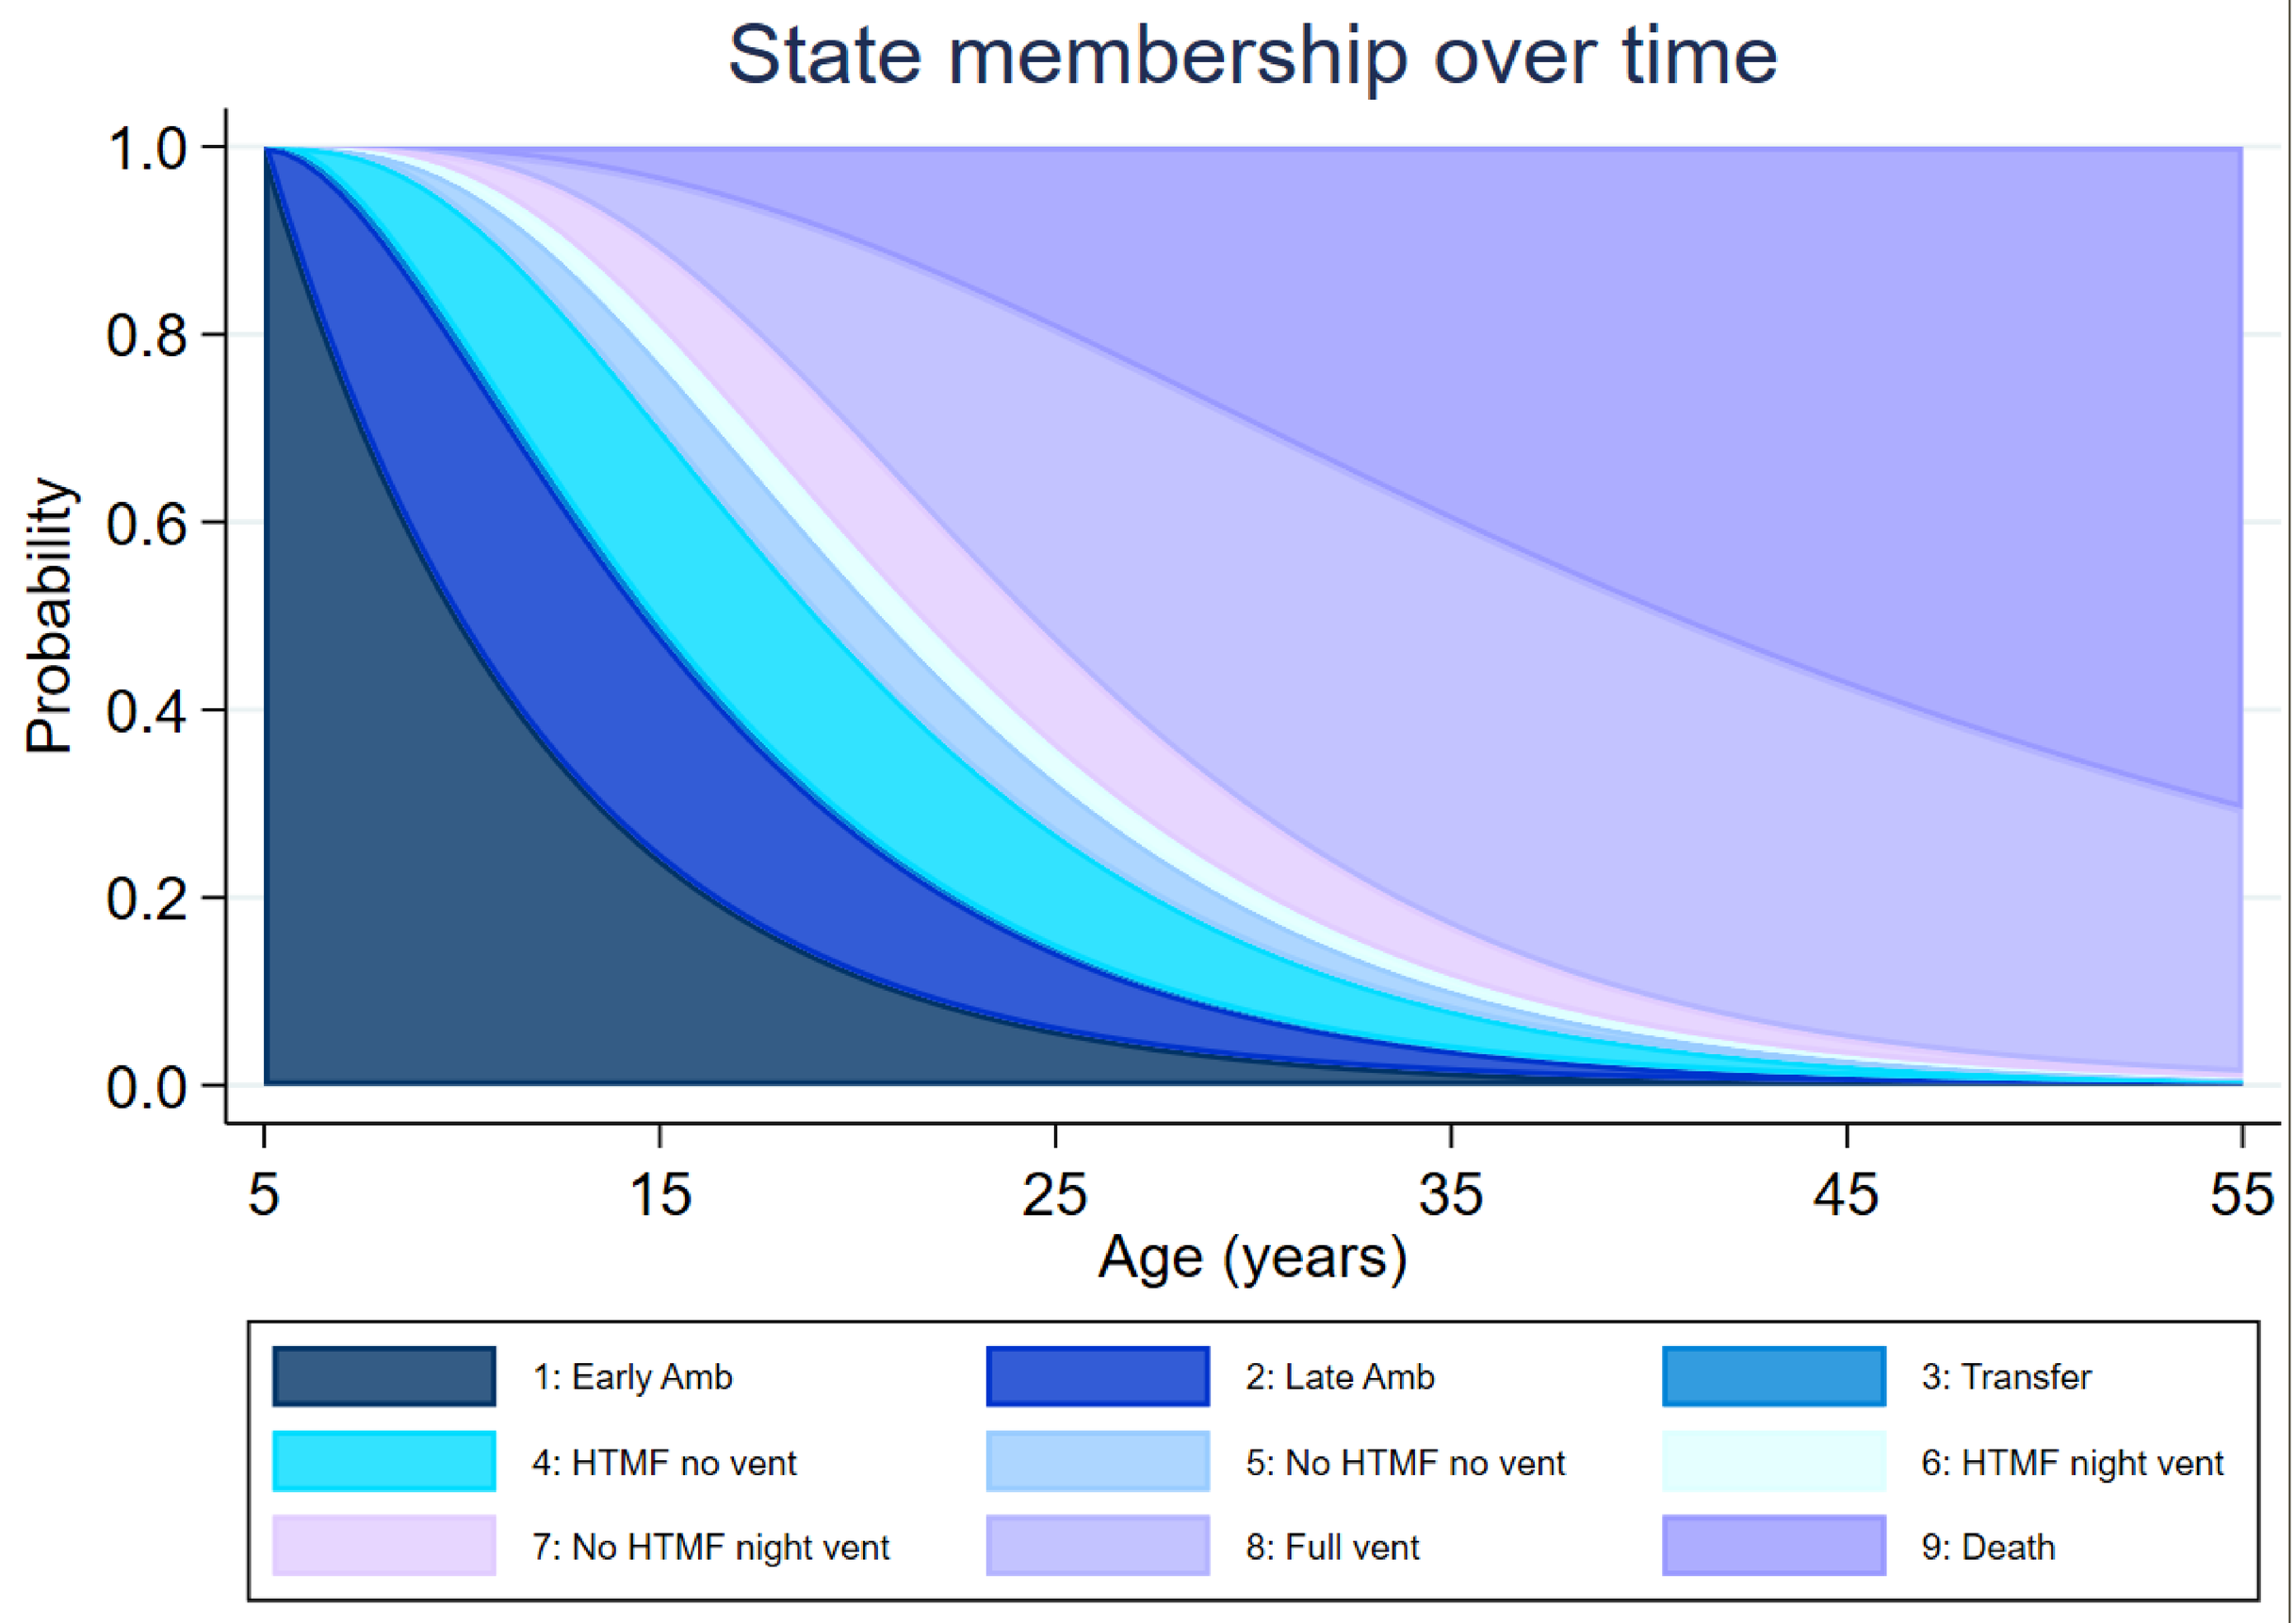
State membership over time
Probability
1.0
0.8
0.6
0.4
0.2
0.0
5
15
25
35
45
55
Age (years)
1: Early Amb
2: Late Amb
3: Transfer
4: HTMF no vent
5: No HTMF no vent
6: HTMF night vent
7: No HTMF night vent
8: Full vent
9: Death

Figure 2: State occupancy probabilities for DMD.

Results

1005 patients from 11 studies were available for intermediate disease transitions. The average age a patient progressed to each health state was consistent with clinical opinion. Median survival of patients with DMD was estimated to be 40 years (see Figure 2), assuming DMD diagnosis at age 5. A longer length of stay in state 8 was estimated than prior belief expected, likely due to the clinical trial data (informing the intermediate transitions) and mortality data coming from different cohorts. Analysis on steroid users suggested longer times spent in later states, with slower transition rates.

Conclusion

Natural history model discussion

This study models the full natural history of DMD in a single unified analysis and highlights some of the challenges of working in a rare disease area. The model quantified vital measures such as length of stay in each state and life expectancy, which can readily inform future economic decision models. New treatments targeted at health states of DMD can be integrated into the natural history model.

Future work

The extracted mortality data will be supplemented by data from the Clinical Practice Research Datalink (CPRD), an electronic health record containing survival data on over 600 patients with DMD. Further elicitation will also be conducted on transitions in and out of state 3 to investigate and reduce uncertainty about the model fit. This study identifies the need to focus future data collection around the transfer state in clinical studies.

References

[1] Criritcal Path Institute, Tucson, AZ. Available at: https://c-path.org/programs/d-rsc/
[2] Guyot P, Ades AE, Ouwens JNM, Welton NJ. *BMC Med. Res. Meth. 2012; 12:9*

Acknowledgements

- The presenting author, Jonathan Broomfield, declares no real or perceived conflicts of interest in relation to this presentation.
- This project is funded by Duchenne UK, Catabasis Pharmaceuticals Inc, Pfizer Inc, PTC Therapeutics, Roche, Sarepta Therapeutics Inc, Solid Biosciences, Santhera Pharmaceuticals Holding AG, Wave Lifesciences USA Inc,

Affiliations

1. University of Leicester, Leicester, UK
2. Karolinska Institutet, Stockholm, SWE
3. Critical Path Institute, Tucson, AZ, USA
4. Translational and Clinical Research Institute, Newcastle University and Newcastle Hospitals NHS Foundation Trust, UK
5. Project HERCULES, Duchenne UK, London, UK