Insights into generalized pustular psoriasis (GPP) using real-world data

Kotowsky N[1], Gao R[1], Singer D[1], Garry EM[2], Golembesky AK[3]

[1]Boehringer Ingelheim Pharmaceuticals, Inc., Ridgefield, CT, USA; [2]Aetion, Inc., Boston, MA, USA;
[3]Boehringer Ingelheim International GmbH, Ingelheim, Germany

Summary

- This retrospective cohort study (using Optum® Clinformatics™ Data Mart, a US administrative claims database) aimed to examine the demographics, comorbidities, use of concurrent medication and hospitalisation rates for patients with GPP compared with those with psoriasis vulgaris (PsO) in a real-world setting in the USA
- Patients with GPP had numerically higher amounts of comorbidities and differing concomitant medication burden compared with those with PsO, reiterating that patients with GPP have a different clinical profile to patients with PsO
- Patients with GPP more often had hospital visits for their condition and associated comorbidities than the non-psoriasis group
- To date, there is little real-world evidence characterising patients with GPP; this analysis suggests that patients with GPP have more healthcare resource utilisation needs than those in a matched, non-psoriasis cohort, as well as differing needs to patients with PsO, highlighting an unmet need among patients with GPP

Background

- Generalized pustular psoriasis (GPP) is a rare, severe and potentially life-threatening disease characterised by recurrent acute flares that consist of a disseminated erythematous skin rash with sterile pustules, causing significant morbidity and in some cases, mortality[1–9]
 - There are a number of associated comorbidities, such as metabolic (hypertension, hyperlipidaemia, diabetes), hepatic (fatty liver), cardiac (ischaemic heart disease), respiratory (asthma, tuberculosis) and neurological (Parkinson's disease, cerebrovascular accident) disorders[4]
 - Reported mortality rates range between 2% and 25%[4–7]
- The clinical, pathological and genetic features of GPP establish it as a distinct disease entity from psoriasis vulgaris (PsO)[2,9–11]
- In the USA, there are currently no approved biologic treatments for GPP, unlike PsO
- To date, there is little real-world evidence characterising patients with GPP. By understanding the burden of disease in this population, targeted interventions that improve patient quality of life can be developed
- This study aimed to examine the demographics, comorbidities, use of concurrent medication and hospitalisation rates for patients with GPP compared with those with PsO in a real-world setting in the USA

Methods

- A retrospective cohort study was conducted using Optum® Clinformatics™ Data Mart, a US administrative claims database[12] **(Figure 1)**
- All analyses were conducted using the Aetion Evidence Platform®, a scientifically validated analytic platform
- Inclusion criteria:
 - Patients with GPP or PsO were identified if they had at least one inpatient or two outpatient ICD-10 (International Classification of Disease, tenth revision) code(s) (L40.1 or L40.0 diagnosis codes, respectively), separated by at least 30 days but within 365 days of one another
 - Patients must have had 180 days (6 months) of continuous baseline enrolment prior to study entry
- Patient selection started on 1 October 2015
- The study period was from 1 April 2016 to 31 March 2019 **(Figure 1)**
- Patients were followed up for a minimum of 12 months after cohort entry

Figure 1. Study design

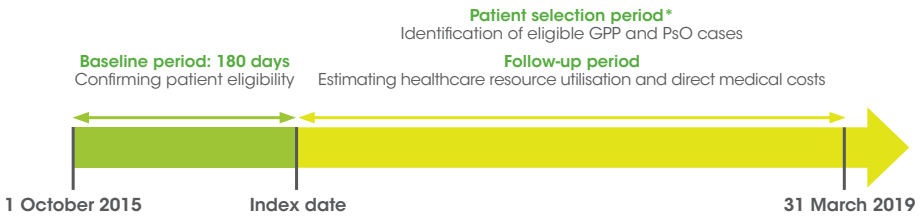
Patient selection period*
Identification of eligible GPP and PsO cases
Baseline period: 180 days
Confirming patient eligibility
Follow-up period
Estimating healthcare resource utilisation and direct medical costs
1 October 2015
Index date
31 March 2019

*Patients entered a cohort when they fulfilled all study eligibility criteria.
GPP, generalized pustular psoriasis; PsO, psoriasis vulgaris.

- Patients with GPP were matched to a non-psoriasis (excludes any psoriasis; ICD-10 code: L40.x) comparator 1:4, based on age and sex, and were also compared with a cohort of patients with PsO
- The following outcomes were evaluated:
 1. Comorbidities at baseline, including (but not limited to) hormonal/metabolic conditions, pulmonary conditions and psychiatric conditions
 2. Concomitant medications at baseline for the treatment of comorbidities
 3. Dermatology treatments received during the 12 months of follow-up
 4. Healthcare resource utilisation, including outpatient, inpatient and emergency department (ED) visits during the follow-up period
- No formal statistical analysis was undertaken and only descriptive statistics were performed

Results

Patient population and demographics

- In total, 1669 patients with GPP and 60,419 with PsO were identified at baseline, with 1014 patients with GPP and 32,665 with PsO completing 12 months of follow-up **(Figure 2)**

Figure 2. Study population

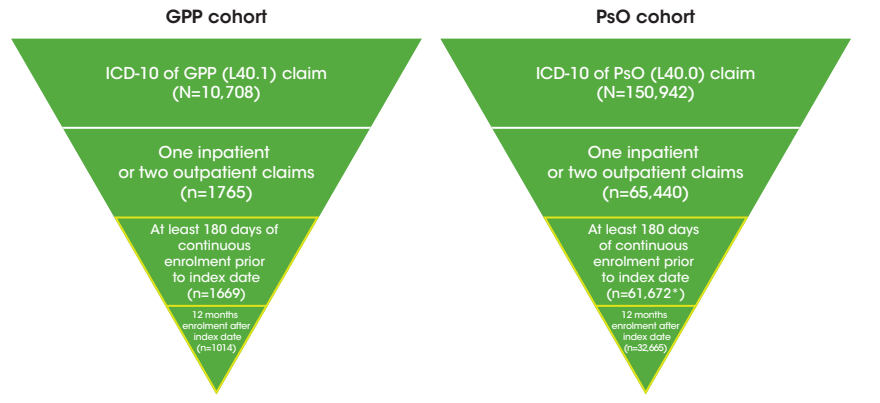
GPP cohort
ICD-10 of GPP (L40.1) claim
(N=10,708)
One inpatient
or two outpatient claims
(n=1765)
At least 180 days of continuous enrolment prior to index date
(n=1669)
12 months enrolment after index date
(n=1014)
PsO cohort
ICD-10 of PsO (L40.0) claim
(N=150,942)
One inpatient
or two outpatient claims
(n=65,440)
At least 180 days of continuous enrolment prior to index date
(n=61,672*)
12 months enrolment after index date
(n=32,665)

*1253 patients with PsO were excluded due to the presence of either GPP or PPP on or prior to cohort entry. Total number of patients with PsO at baseline was 60,419.
GPP, generalized pustular psoriasis; ICD-10, international classification of diseases, tenth revision; PsO, psoriasis vulgaris.

- Patients with GPP tended to be female (67.6%) and had a mean age of 63.9 years, whereas 50.7% of patients with PsO were female, with a mean age of 56.4 years **(Table 1)**

Table 1. Patient demographics at baseline

	GPP n=1669	PsO n=60,419	Non-psoriasis matched cohort n=6676
Female, n (%)	1128 (67.6)	30,621 (50.7)	4512 (67.6)
Age, years, mean (SD)	63.9 (15.2)	56.4 (17.3)	59.6 (19.9)
<18 years, n (%)	17 (1.0)	1135 (1.9)	68 (1.0)
18–64 years, n (%)	739 (44.3)	37,645 (62.3)	2956 (44.3)
≥65 years, n (%)	913 (54.7)	21,637 (35.8)	3652 (54.7)
Commercial payers, n (%)	580 (34.8)	37,774 (62.5)	3127 (46.8)
Medicare payers, n (%)	1089 (65.2)	22,645 (37.5)	3549 (53.2)

GPP, generalized pustular psoriasis; PsO, psoriasis vulgaris; SD, standard deviation.

Comorbidities

- Patients with GPP, compared with patients with PsO and the matched comparators in the non-psoriasis group, were more likely to suffer from psoriatic arthritis (GPP: 11.8%; PsO: 7.0%; non-psoriasis: 0.1%), anxiety (11.6%; 6.7%; 5.8%) and depression (10.7%; 5.5%; 5.0%) at baseline **(Figure 3)**
- Patients with GPP were also more likely to suffer from hyperlipidaemia, obesity and COPD than patients with PsO and the non-psoriasis group at baseline **(Figure 3)**

Figure 3. Proportion of patients with comorbidities at baseline

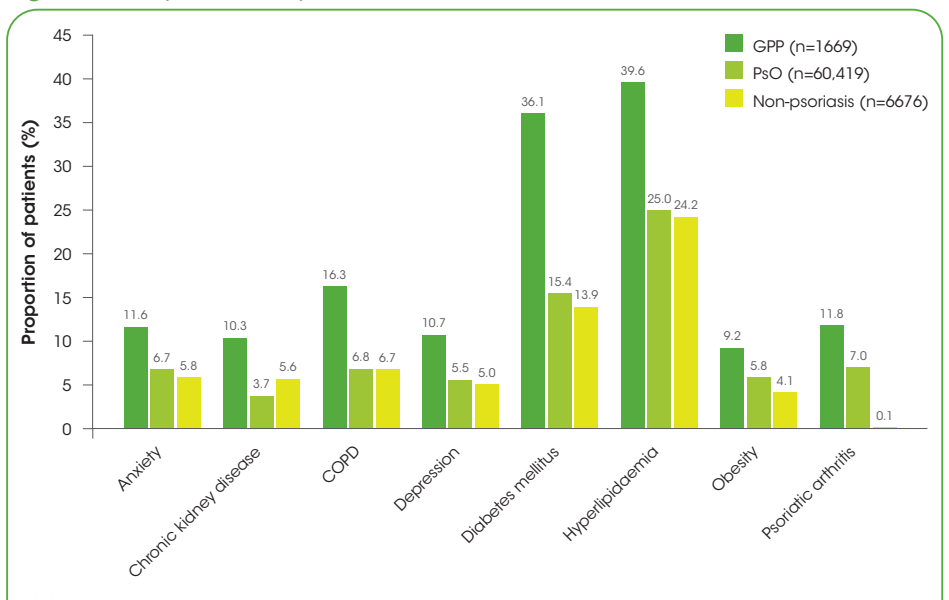
GPP (n=1669)
PsO (n=60,419)
Non-psoriasis (n=6676)
Proportion of patients (%)
45
40
35
30
25
20
15
10
5
0
Anxiety 11.6 6.7 5.8
Chronic kidney disease 10.3 3.7 5.6
COPD 16.3 6.8 6.7
Depression 10.7 5.5 5.0
Diabetes mellitus 36.1 15.4 13.9
Hyperlipidaemia 39.6 25.0 24.2
Obesity 9.2 5.8 4.1
Psoriatic arthritis 11.8 7.0 0.1

COPD, chronic obstructive pulmonary disease; GPP, generalized pustular psoriasis; PsO, psoriasis vulgaris.

Medication use

- Of the 1014 GPP patients with 12 months of follow-up, 449 (44.3%) were treated with a systemic therapy during follow-up, including:
 - 51 (5.0%) who were treated with a biologic only
 - 153 (15.0%) who were treated with a non-biologic systemic therapy only
- More than one-third (35.6%) of patients with GPP were treated with a topical steroid medication, 19.3% were treated with a topical non-steroid medication and 27.9% with systemic steroids **(Figure 4)**
 - The use of systemic steroids was similar in patients with GPP (27.9%) and those with PsO (25.2%); however, the use of topical steroids was more common in patients with PsO (55.8%) than in those with GPP (35.6%)
 - Medications were not mutually exclusive as patients could have been treated with both topical medications and systemic steroids during the 12-month follow-up
- Methotrexate was the most commonly used oral systemic therapy (10.8%) in patients with GPP, while adalimumab and ustekinumab were the most commonly used biologics (6.7% and 4.8%, respectively) **(Figure 4)**

Figure 4. Medication use in patients with GPP and with PsO during 12-months' follow-up

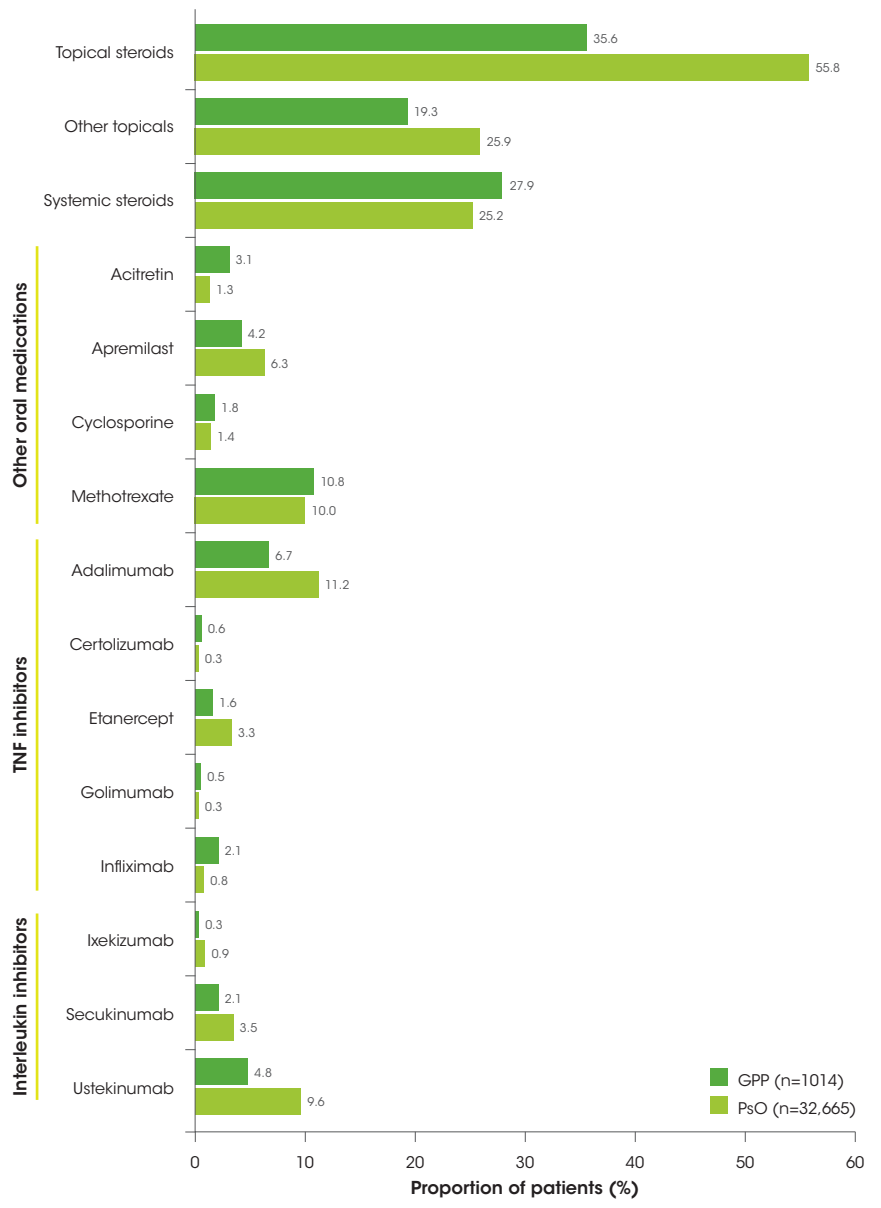
Topical steroids 35.6 55.8
Other topicals 19.3 25.9
Systemic steroids 27.9 25.2
Other oral medications
Acitretin 3.1 1.3
Apremilast 4.2 6.3
Cyclosporine 1.8 1.4
Methotrexate 10.8 10.0
TNF inhibitors
Adalimumab 6.7 11.2
Certolizumab 0.6 0.3
Etanercept 1.6 3.3
Golimumab 0.5 0.3
Infliximab 2.1 0.8
Interleukin inhibitors
Ixekizumab 0.3 0.9
Secukinumab 2.1 3.5
Ustekinumab 4.8 9.6
GPP (n=1014)
PsO (n=32,665)
0
10
20
30
40
50
60
Proportion of patients (%)

GPP, generalized pustular psoriasis; PsO, psoriasis vulgaris; TNF, tumour necrosis factor.

Outpatient, inpatient and ED visits during the 12-month follow-up

- Compared with the non-psoriasis group, patients with GPP had a higher total number of outpatient visits per patient (median: 21 vs 9, mean: 28.7 vs 15.3)
- A higher proportion of patients with GPP had more inpatient visits (22.5% vs 10.4%), with the mean number of inpatient visits for patients with GPP being 0.5 and the non-psoriasis group being 0.2
 - The duration of inpatient stays for patients with GPP tended to be longer than in the matched non-psoriasis group (median: 8 vs 5 days, mean 16.8 vs 14.4 days)
- Patients with GPP and patients in the non-psoriasis cohort both had a median total number of visits to the ED of 0, with the mean number of visits being 2.2 and 0.8, respectively
- However, a substantially higher proportion of patients with GPP had an ED visit than those in the non-psoriasis group (40.5% vs 23.0%)
- At baseline, concomitant medication burden shows that the most common prescription in patients with GPP were antihypertensives (58.8%)
 - Antihypertensives were also the most common prescription for patients with PsO; however, the proportion of patients with a prescription was much lower (40.8%)
 - The proportion of patients with GPP needing opioid pain medication was higher than patients with PsO (29.9% vs 15.8%)

Table 2. Concomitant medications at baseline

Medication, n (%)	GPP n=1669	PsO n=60,419
Antihypertensives	981 (58.8)	24,648 (40.8)
Statins	674 (40.4)	16,102 (26.7)
Antibiotics	635 (38.0)	20,112 (33.3)
Type 2 diabetes medication	420 (25.2)	7483 (12.4)
Asthma medication	482 (28.9)	11,331 (18.8)
COPD medication	285 (17.1)	6133 (10.2)
Psychiatric medication	679 (40.7)	17,428 (28.8)
Opioid pain medication	499 (29.9)	9571 (15.8)
Sleep medication (non-benzodiazepine)	51 (3.1)	1957 (3.2)

COPD, chronic obstructive pulmonary disease; GPP, generalized pustular psoriasis; PsO, psoriasis vulgaris.

Conclusions

- This study summarises the clinical characteristics of 1669 patients with GPP in a real-world setting in the USA
- This study showed that patients with GPP have numerically higher amounts of comorbidities and differing concomitant medication burden compared with those with PsO, reiterating that patients with GPP have a different clinical profile to patients with PsO
- Despite differences in clinical characteristics, GPP patients were treated similarly to those with PsO
- Patients with GPP had numerically higher healthcare visits than those with PsO and patients matched from the general population
 - Together, these data highlight that patients with GPP have higher HCRU than patients with PsO and the non-psoriasis cohort, which may represent more severe disease
- The study used data from an administrative claims database; these data were collected for reimbursement purposes rather than scientific research, thus, there may be miscoding
 - Data on treatments are generally considered to be high quality, reflecting what the patient received at the pharmacy. Biologics were identified by both the Healthcare Common Procedure Coding System and National Drug Codes
- This analysis suggests that patients with GPP have more healthcare resource utilisation needs than those in the matched, non-psoriasis cohort, as well as differing needs to those in the PsO patient population, highlighting an unmet need among patients with GPP

References

1. Bachelez H. *Br J Dermatol* 2018;178:614–618
2. Navarini AA, et al. *J Eur Acad Dermatol Venereol* 2017;31:1792–1799
3. Gooderham MJ, et al. *Expert Rev Clin Immunol* 2019;15:907–919
4. Choon SE, et al. *Int J Dermatol* 2014;53:676–684
5. Ryan TJ & Baker H. *Br J Dermatol* 1971;85:407–411
6. Zelickson BD & Muller SA. *Arch Dermatol* 1991;127:1339–1345
7. Augey F, et al. *Eur J Dermatol* 2006;16:669–673
8. Jin H, et al. *J Dermatol* 2015;42:674–678
9. Johnston A, et al. *J Allergy Clin Immunol* 2017;140:109–120
10. Furue K, et al. *Acta Derm Venereol* 2018;98:5–13
11. Twelves S, et al. *J Allergy Clin Immunol* 2019;143:1021–1026
12. Optum Clinformatics Data Mart 2017. https://www.optum.com/content/dam/optum/resources/productSheets/Clinformatics_for_Data_Mart.pdf [Accessed 17 March 2020]

Disclosures

This study was funded by Boehringer Ingelheim. N Kotowsky, D Singer, R Gao and AK Golembesky are full-time employees of Boehringer Ingelheim. EM Garry is employed by Aetion, contracted by Boehringer Ingelheim.

Acknowledgements

Editorial support was provided by Amy Pashler, PhD from OPEN Health Medical Communications (London, UK) and was funded by Boehringer Ingelheim.

The 10th European Conference on Rare Diseases.

Corresponding author: Amanda K Golembesky

Simply click the icon to access an interactive microsite for this Smart poster