Burden of Illness and Quality of Life Among Patients with Tuberous Sclerosis Complex: Assessed as Part of the International TOSCA Study

Anna Jansen*[1], Elena Belousova[2], Mirjana P. Benedik[3], Tom Carter[4], Vincent Cottin[5], Paolo Curatolo[6], Maria Dahlin[7], Lisa D´Amato[8], Guillaume B. d'Augères[9], Petrus J. de Vries[10], José C. Ferreira[11], Martha Feucht[12], Carla Fladrowski[13,14], Christoph Hertzberg[15], Sergiusz Jozwiak[16,17], John A. Lawson[18], Alfons Macaya[19], Ruben Marques[8,20], Rima Nabbout[21], Finbar O'Callaghan[22], Jiong Qin[23], Valentin Sander[24], Matthias Sauter[25], Seema Shah[26], Yukitoshi Takahashi[27], Renaud Touraine[28], Sotiris Youroukos[29], Bernard Zonnenberg[30], Chris J Kingswood[31] on behalf of TOSCA Consortium and TOSCA Investigators

[1]Pediatric Neurology Unit, Department of Pediatrics, UZ Brussel VUB, Brussels, Belgium; [2]Research and Clinical Institute of Pediatrics, Pirogov Russian National Research Medical University, Moscow, Russian Federation; [3]SPS Pediatrična Klinika, Ljubljana, Slovenia; [4]TSA Tuberous Sclerosis Association, Nottingham, United Kingdom; [5]Hôpital Louis Pradel, Claude Bernard University Lyon 1, Lyon, France; [6]Tor Vergata University Hospital, Rome, Italy; [7]Karolinska University Hospital, Stockholm, Sweden; [8]Novartis Farma S.p.A., Origgio, Italy; [9]Association Sclérose Tubéreuse de Bourneville, Gradignan, France; [10]Division of Child and Adolescent Psychiatry, University of Cape Town, Cape Town, South Africa; [11]Centro Hospitalar Lisboa Ocidental, Lisbon, Portugal; [12]Universitätsklinik für Kinder-und Jugendheilkunde, Vienna, Austria; [13]Associazione Sclerosi Tuberosa ONLUS, Milan, Italy; [14]European Tuberous Sclerosis Complex Association, In den Birken, Dattein, Germany; [15]Vivantes-Klinikum Neukölln, Berlin, Germany; [16]Department of Child Neurology, Warsaw Medical University, Warsaw, Poland; [17]Department of Neurology and Epileptology, The Children's Memorial Health Institute, Warsaw, Poland; [18]The Tuberous Sclerosis Multidisciplinary Management Clinic, Sydney Children's Hospital, Randwick, NSW, Australia; [19]Hospital Universitari Vall d'Hebron, Barcelona, Spain; [20]Institute of Biomedicine University of Leon, Spain; [21]Department of Pediatric Neurology, Necker Enfants Malades Hospital, Paris Descartes University, Paris, France; [22]Institute of Child Health, University College London, London, United Kingdom; [23]Department of Pediatrics, Peking University People's Hospital (PKUPH), Beijing, China; [24]Tallinn Children Hospital, Tallinn, Estonia; [25]Klinikverbund Kempten-Oberallgäu gGmbH, Kempten, Germany; [26]Novartis Healthcare Pvt. Ltd., Hyderabad, India; [27]National Epilepsy Center, Shizuoka Institute of Epilepsy and Neurological Disorders, NHO, 886 Urushiyama Aoi-ku Shizuoka, Japan; [28]Department of Genetics, CHU-Hôpital Nord, Saint Etienne, France; [29]St. Sophia Children's Hospital, Athens, Greece; [30]University Medical Center, Utrecht, Netherlands; [31]Sussex Kidney Unit, Royal Sussex County Hospital, Eastern Road, Brighton BN2 5BE, United Kingdom

Background

- Tuberous sclerosis complex (TSC) is a multisystem genetic disorder with a global incidence of 1 per 6-10,000 live births (1.5 million people).[1,2]
- Although rare, the presentation of TSC manifestations are complex with severity varying between patients.[1,3-5]
- TSC is characterized by growth of benign tumours (hamartomas) in various organs throughout the body, including the brain, kidney, lungs, and skin.[1]
 - It also causes TSC-Associated Neuropsychiatric Disorders (TAND), having a strong impact on the quality of life (QoL) and psychosocial status of both the patients and their families.[6,7]
- Past research on the TSC mainly focused on the epidemiology, diagnostic criteria and the treatment efficacy[8]; there is no large-scale study evaluating the disease implications on QoL and social health of TSC patients and their families.
- Here we present the results of the **T**uber**O**us **SC**lerosis registry to increase disease **A**wareness (TOSCA) study research project that aimed to evaluate the impact of TSC on the lives of patients and/or their caregivers in terms of burden of illness (BOI) and QoL.

Methods

- TOSCA was a large-scale, non-interventional study designed with a "core" section to record patient background information, 6 research projects that focused on specific disease manifestations, and a European post authorization safety study (PASS).
- Patients from 7 European countries who were enrolled into the TOSCA study were also eligible for this QoL research project after signing specific informed consent.
- All enrolled patients or their caregivers were asked to complete validated paper questionnaires in the local languages to collect information on BOI and QoL:
 - EuroQol-5D (EQ-5D) for adult patients (EQ-5D proxy version 1 was completed by the caregiver for children and for adults who were unable to complete the report by themselves
 - Quality of Life in Epilepsy Inventory-31-Problems (QOLIE-31P) for adult patients with epilepsy
 - Life in Childhood Epilepsy (QOLCE) for children <10 years old with epilepsy (filled by caregivers)
 - Quality of Life in Epilepsy Inventory for Adolescents-48 (QOLIE-AD-48) for children aged 11-17 years with epilepsy
- For BOI, data were collected based on ancillary questions developed in collaboration with patient representatives and aimed to address specific disease topics including patient assistance requirements and support/rights, allowance and insurance, access to healthcare resources, sources of information, clinical trial experience, satisfaction with treatment aspects, impact on education and employment situation of the individual with TSC and caregivers or other family members, financial impact, and genetic testing and counseling. In addition, all physicians were requested to answer questions in PRF (Physician Reported Form) regarding hospitalization and consultations in the past 12 months.

Results

- A total of 143 patients (55 adults and 88 children) from 7 European countries were enrolled in this research project as part of the TOSCA study (**Table 1**).
 - Among the 55 adults, 38 were able to complete the questionnaire independently, whereas caregivers completed the questionnaire for 17 patients who were unable to complete the questionnaires by themselves.
 - Among the 88 children, 17 were able to complete the questionnaire independently, whereas caregivers completed the questionnaire for 71 patients who were unable to complete them.

Table 1. Demographic Characteristics

		Adults (≥18 years) (N=55)	Children (<18 years) (N=88)	Overall (N=143)
Sex, n (%)	Male	17 (30.9)	37 (42.0)	54 (37.8)
	Female	38 (69.1)	50 (56.8)	88 (61.5)
	Missing	0	1 (1.1)	1 (0.7)
Age at consent (years)	n	55	87	142
	Mean (SD)	35.6 (12.71)	9.8 (4.30)	19.8 (15.24)
	Median (range)	34 (18-72)	11 (3-20)	14 (3-72)
Duration of TSC (years)	n	54	87	141
	Mean (SD)	21.3 (10.33)	8.7 (4.25)	13.5 (9.44)
	Median (range)	19.6 (1.8-43.5)	8.6 (1.6-19.9)	11.2 (1.6-43.5)
Country, n (%)	Belgium	13 (23.6)	11 (12.5)	24 (16.8)
	France	9 (16.4)	21 (23.9)	30 (21.0)
	Germany	0	11 (12.5)	11 (7.7)
	Italy	30 (54.5)	28 (31.8)	58 (40.6)
	Spain	0	11 (12.5)	11 (7.7)
	Sweden	0	6 (6.8)	6 (4.2)
	UK	3 (5.5)	0	3 (2.1)
Duration of epilepsy (years) at start of research project	Patients with epilepsy	28	39	67
	n	27	39	66
	Mean (SD)	27.1 (13.23)	9.3 (4.10)	16.6 (12.53)
	Median (range)	31.2 (2.7-55.4)	9.4 (2.8-16.9)	12.8 (2.7-55.4)
Epilepsy types*, n (%)	Infantile spasms (IS)	7 (25.0)	26 (66.7)	33 (49.3)
	Focal seizures (FS)	24 (85.7)	34 (87.2)	58 (86.6)
	Both IS and FS	5 (17.9)	23 (59.0)	28 (41.8)
	Other	11 (39.3)	13 (33.3)	24 (35.8)

*Includes a patient with more than 1 seizure type.
FS, focal seizure; IS, infantile spasm; SD, standard deviation; TSC, tuberous sclerosis complex; UK, United Kingdom

Burden of TSC

Social and healthcare needs

- In total, 14 of 55 adults (25.5%) and 21 of 88 of children (23.9%) needed extra assistance at home. Of these, 85.7% of adults and 76.2% of children, assistance was provided by unpaid caregivers (a family member or friend) (**Table 2**).
- Seventy-three patients (51%) reported that social services did not give adequate support and information.
- Forty-one patients (28.7%) reported that they did not have any health insurance.

Table 2. Burden of Illness

			Adults (≥18 years) (N=55)	Children (<18 years) (N=88)	Overall (N=143)
Assistance at home, n (%)			14 (25.5)	21 (23.9)	35 (24.5)
Allowance, n (%)	Disability allowance	Yes	21 (38.2)	45 (51.1)	66 (46.2)
		No	33 (60.0)	38 (43.2)	71 (49.7)
		Missing	1 (1.8)	5 (5.7)	6 (4.2)
	Caregiver allowance	Yes	0	10 (11.4)	10 (7.0)
		No	46 (83.6)	68 (77.3)	114 (79.7)
		Missing	9 (16.4)	10 (11.4)	19 (13.3)
	Psychological counseling	Yes	3 (5.5)	12 (13.6)	15 (10.5)
		No	43 (78.2)	67 (76.1)	110 (76.9)
		Missing	9 (16.4)	9 (10.2)	18 (12.6)
	Social worker assistance	Yes	1 (1.8)	7 (8.0)	8 (5.6)
		No	44 (80.0)	71 (80.7)	115 (80.4)
		Missing	10 (18.2)	10 (11.4)	20 (14.0)
	Social services support	Yes	2 (3.6)	4 (4.5)	6 (4.2)
		No	43 (78.2)	74 (84.1)	117 (81.8)
		Missing	10 (18.2)	10 (11.4)	20 (14.0)
	Support in completing paperwork regarding benefits available	Yes	2 (3.6)	5 (5.7)	7 (4.9)
		No	42 (76.4)	75 (85.2)	117 (81.8)
		Missing	11 (20.0)	8 (9.1)	19 (13.3)
Insurance, n (%)	Yes		22 (40.0)	61 (69.3)	83 (58.0)
	No		23 (41.8)	18 (20.5)	41 (28.7)
	Not applicable		2 (3.6)	0	2 (1.4)
	Missing		8 (14.5)	9 (10.2)	17 (11.9)
Genetic testing, n (%)	Patient had genetic testing for TSC		47 (85.5)	70 (79.5)	117 (81.8)
	Patient was offered genetic testing but did not do it		1 (1.8)	4 (4.5)	5 (3.5)
	Patient had not been offered genetic testing for TSC		4 (7.3)	7 (8.0)	11 (7.7)
Genetic counseling, n (%)	Patient had genetic counseling		36 (65.5)	52 (59.1)	88 (61.5)
	Patient was offered genetic counseling but decided not to have it		0	3 (3.4)	3 (2.1)
	Patient had not been offered genetic counseling for TSC		10 (18.2)	23 (26.1)	33 (23.1)
Number of doctors managing TSC, n (%)	1		18 (32.7)	25 (28.4)	43 (30.1)
	2		6 (10.9)	14 (15.9)	20 (14.0)
	3		5 (9.1)	11 (12.5)	16 (11.2)
	>3		22 (40.0)	37 (42.0)	59 (41.3)
	Missing		4 (7.3)	1 (1.1)	5 (3.5)
TSC is managed by*, n (%)	General practitioner/family doctor		15 (27.3)	18 (20.5)	33 (23.1)
	TSC specialist		44 (80.0)	51 (58.0)	95 (66.4)
	Other specialist		26 (47.3)	56 (63.6)	82 (57.3)
	Missing		1 (1.8)	2 (2.3)	3 (2.1)
Access to TSC clinic when required, n (%)	Yes		45 (81.8)	56 (63.6)	101 (70.6)
	No		7 (12.7)	27 (30.7)	34 (23.8)
	Missing		3 (5.5)	5 (5.7)	8 (5.6)

*Participants may have provided more than 1 answer
TSC, tuberous sclerosis complex

- Fifty-nine patients (41.3%) reported that they were followed for TSC managing by more than 3 physicians, whereas TSC was managed by a TSC specialist in 95 patients (66.4%). No access to TSC clinics when required were reported by 34 patients (23.8%), whereas 60 patients (42%) reported that a TSC clinic was > 50 km from their home.
- For 117 patients, genetic testing for TSC was performed (81.8%). However, for 33 patients (23.1%) no genetic counseling was offered.
- Sixty-eight patients (47.6%) were in contact with a TSC association and 69 (48.3%) were not.
- Overall, 65 patients (45.5%) were very satisfied with the level of involvement in treatment decisions, 78 (54.5%) with the explanation of treatment options, 71 (49.7%) with management of all aspects of TSC, 73 (51%) with easy access to the prescribed drugs, 71 (49.7%) with explanations on TSC condition and 63 (44.1%) with medical care for TSC (**Figure 1**).

Figure 1. Satisfaction with Treatment Aspects (N=143)

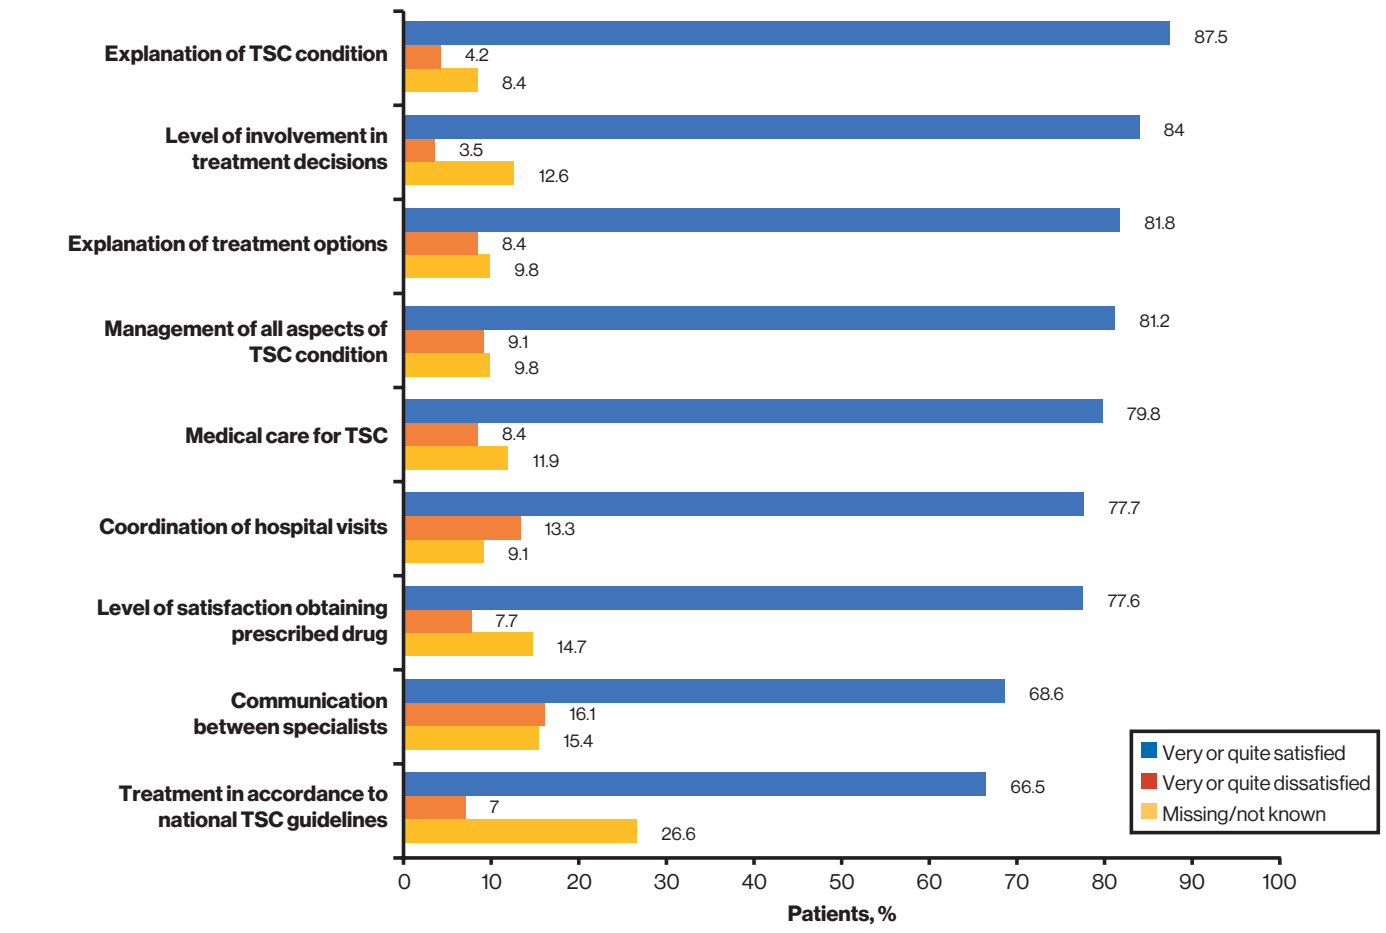
Explanation of TSC condition
87.5
4.2
8.4
Level of involvement in treatment decisions
84
3.5
12.6
Explanation of treatment options
81.8
8.4
9.8
Management of all aspects of TSC condition
81.2
9.1
9.8
Medical care for TSC
79.8
8.4
11.9
Coordination of hospital visits
77.7
13.3
9.1
Level of satisfaction obtaining prescribed drug
77.6
7.7
14.7
Communication between specialists
68.6
16.1
15.4
Treatment in accordance to national TSC guidelines
66.5
7
26.6
Very or quite satisfied
Very or quite dissatisfied
Missing/not known
0
10
20
30
40
50
60
70
80
90
100
Patients, %

TSC, tuberous sclerosis complex

Education (in children) or employment (in adults)

- Of the 88 enrolled children, 57 (64.8%) were in mainstream education. Of these, 37 (42%) received additional support in class.
- Of the 55 enrolled adults, 28 (50.9%) reported that their career/education was impacted by TSC.
- Of the 28 adults with epilepsy, 15 (53.6%) reported an impact on their career/education due to TSC.

Impact on family

- Career/education of other members of the family was impacted by TSC in 15 adults (27.3%). Family, social, and working relationships of the caregiver were impacted by the child's TSC condition in 29 families (52.7%).
- Career/education of the caregiver was impacted by TSC in 50 children (56.8%). Family, social, and working relationships of the caregiver were impacted by the child's TSC condition in 41 families (46.6%).

Pediatric/adult care for adult patients

- Of the 55 enrolled adults, 18 patients (32.7%) reported availability of adult care in their area, and smooth transition from pediatric to adult care was reported in 23 (41.8%).

Physician reported outcomes

- Of the 143 enrolled patients, 42 (29.4%) were hospitalized in the last 12 months since the enrolment in this research project. Main reasons for hospitalization: elective (32 patients [22.4%], mostly due to investigation and nuclear magnetic resonance imaging) and emergency (11 patients, 7.7%, mostly due to epilepsy). In 33 patients (23.1%), the reason for hospitalization was TSC-related.
- With regard to physician consultations in the past 12 months, no TSC-related visits to site were performed in 57 patients (39.9%), no TSC-related visits to other specialists in 42 patients (29.4%) and no TSC-related visits to a primary care physician in 57 patients (39.9%).

Quality of Life

- **EQ-5D questionnaire:** 143 patients filled the EQ-5D questionnaire. From the results of each level of the 5 dimensions, 34 patients (23.8%) reported some difficulty in mobility and 32 (22.4%) reported difficulty in self-care (**Figure 2**). Twenty-six patients (18.2%) were unable to perform usual activities. Sixty-two patients (43.4%) reported moderate anxiety/depression, whereas 6 patients (4.2%) reported extreme anxiety/depression. On the thermometer scale of 0 to 100, (100 being the best state of health imaginable and 0 as the worst state imaginable), the mean score was 70.6.
- **QOLIE-31-P questionnaire:** Twenty-four adult patients filled the questionnaire. The final scale score of the QOLIE-31 questionnaire was 71.6. Most important areas related to epilepsy according to the patient was seizure worry. The mean health state score was 70 (recorded on a scale from 0 to 100, with 0 being the worst health state imaginable and 100 the best).
- **QOLCE questionnaire:** 70 caregivers of patients filled out the questionnaire. Mean final QOLCE questionnaire score was 52.3. The highest score was reported for depression and the lowest for other cognitive issues.

Figure 2. Summary of the EQ-5D Questionnaire

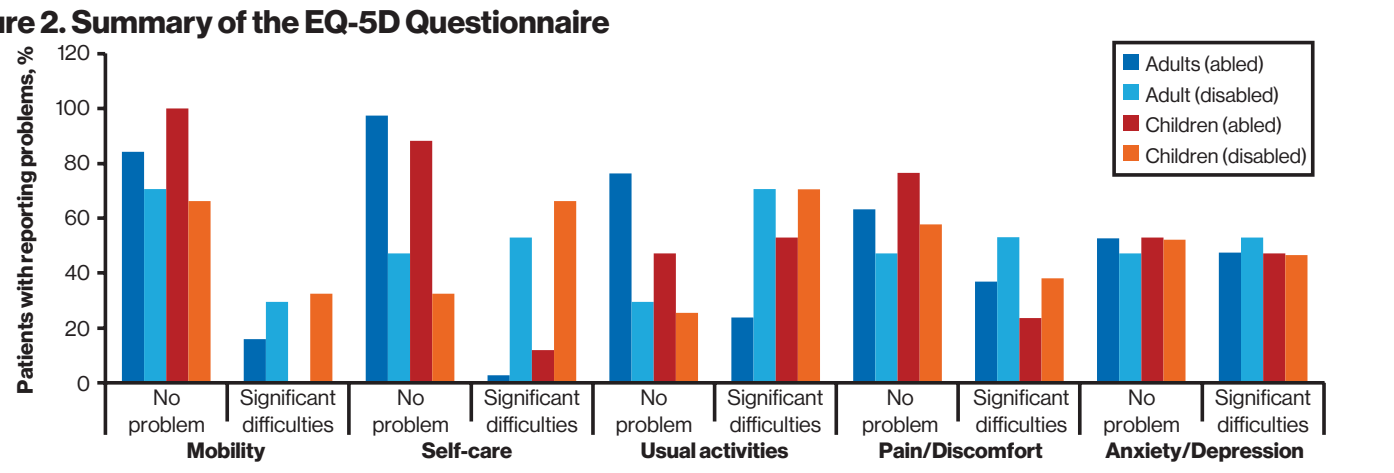
Patients with reporting problems, %
120
100
80
60
40
20
0
No problem
Significant difficulties
Mobility
Self-care
Usual activities
Pain/Discomfort
Anxiety/Depression
Adults (abled)
Adult (disabled)
Children (abled)
Children (disabled)

EQ-5D, EuroQol-5D

Conclusions

- TOSCA is the first multinational clinical study to evaluate BOI and QoL in patients with TSC.
- Overall, the results suggest that although TSC is considered as a rare disease, it has a great impact on the patients and/or their caregivers or family owing to the varied extent and severity of its manifestations among patients.
- TSC has major impacts on patients and their families' health and social well-being. These could be markedly ameliorated by better educational, social, and medical support.

References

1. Curatolo P, et al. Lancet 2008;372:657-668.
2. O'Callaghan FJ, et al. Lancet 1998;351:1490.
3. Kingswood C, et al. Eur J Paediatr Neurol 2016;20:296-308.
4. Staley BA, et al. Pediatrics 2011;127:e117-125.
5. Hallett L, et al. Curr Med Res Opin 2011;27:1571-1583.
6. de Vries PJ, et al. Pediatr Neurol. 2015;52(1):25-35.
7. Krueger DA, et al. Pediatr Neurol 2013;49:255-265.
8. Graffigna G, et al. BMJ open 2013;3:e003707.

Acknowledgments

We would like to thank the patients and patient organisations who took part in the trial and their families and the staff at each site that assisted with the study. We would also like to thank Manojkumar Patel and Pranitha Akula (Novartis Healthcare Pvt. Ltd.) for medical editorial assistance with this poster.

Poster presented at the 10th European Conference for Rare Diseases and Orphan Products; May 14-16, 2020; Virtual.

This study is sponsored by Novartis Pharma AG.

Text: Q45c3e
To: 8NOVA (86682) US Only
+18324604729 North, Central and South Americas; Caribbean; China
+447860024038 UK, a & Russia
+46737494608 Sweden, Europe

Visit the web at:
http://novartis.medicalcongressposters.com/Default.aspx?doc=45c3e

Scan to download a copy of this poster

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.