Aromatic-L-amino acid decarboxylase (AADC) deficiency:
What is it and how is it diagnosed?

Sian O´Neill[1], Christian Werner MD[2], Serene Forte PhD, MPH[3], Axel Boehnke[2]

[1]PTC Therapeutics, Switzerland GmbH, Zug, Switzerland; [2]PTC Therapeutics Germany GmbH, Frankfurt, Germany; [3]PTC Therapeutics Inc, Lynnfield, Massachusetts, USA

AADC deficiency is a severe disease

AADC deficiency typically presents early and can lead to the need for lifelong care, with life-limiting motor and autonomic dysfunction, developmental delay leading to premature death.[1-4]

Patients usually do not develop full head control or the ability to sit unassisted or stand.

AADC deficiency affects patients of any gender, ethnic origin or geographical region.

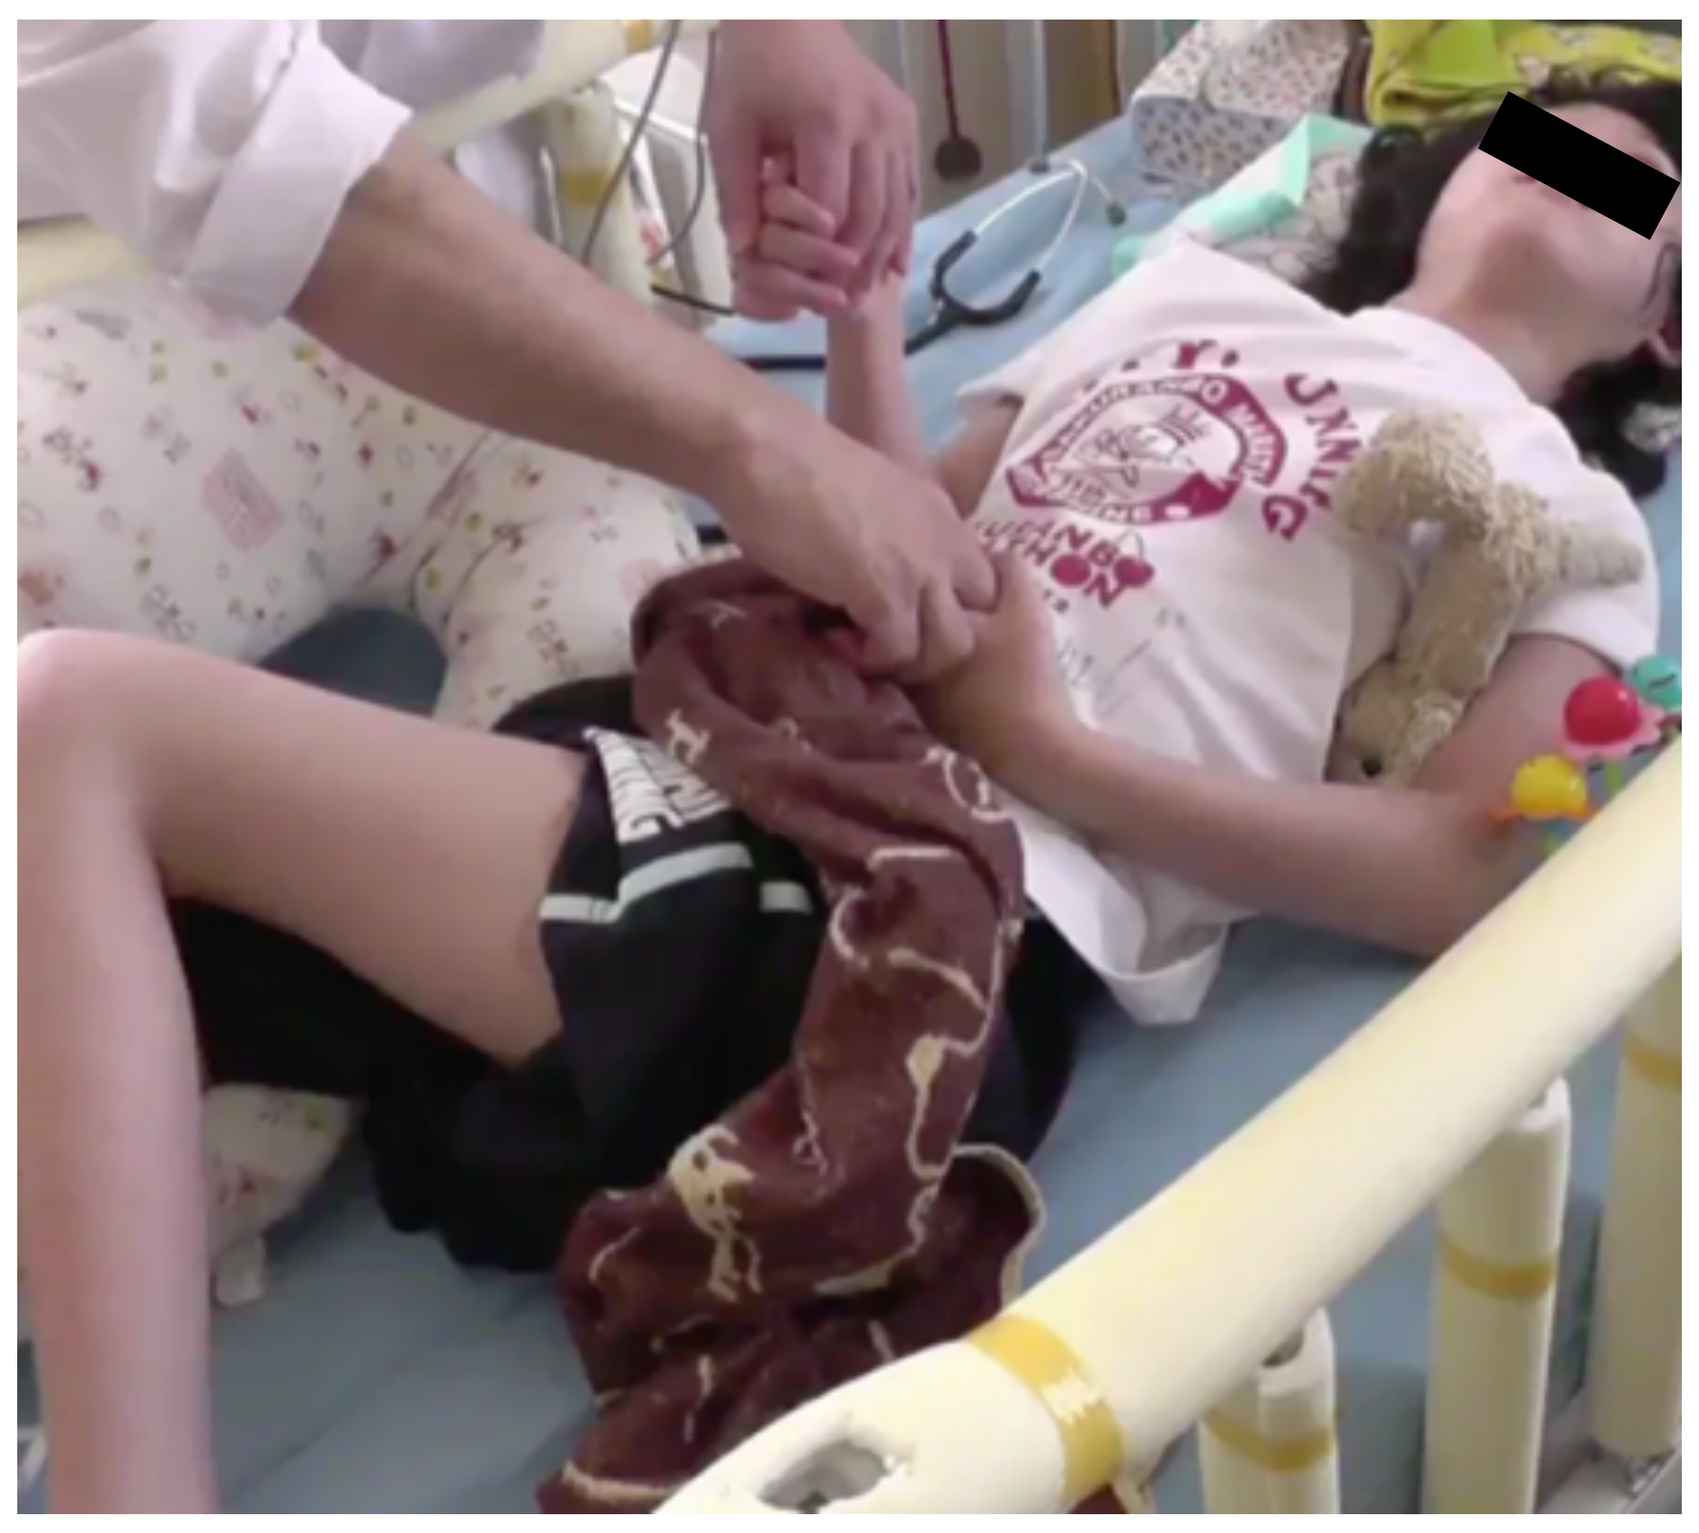

patient 1

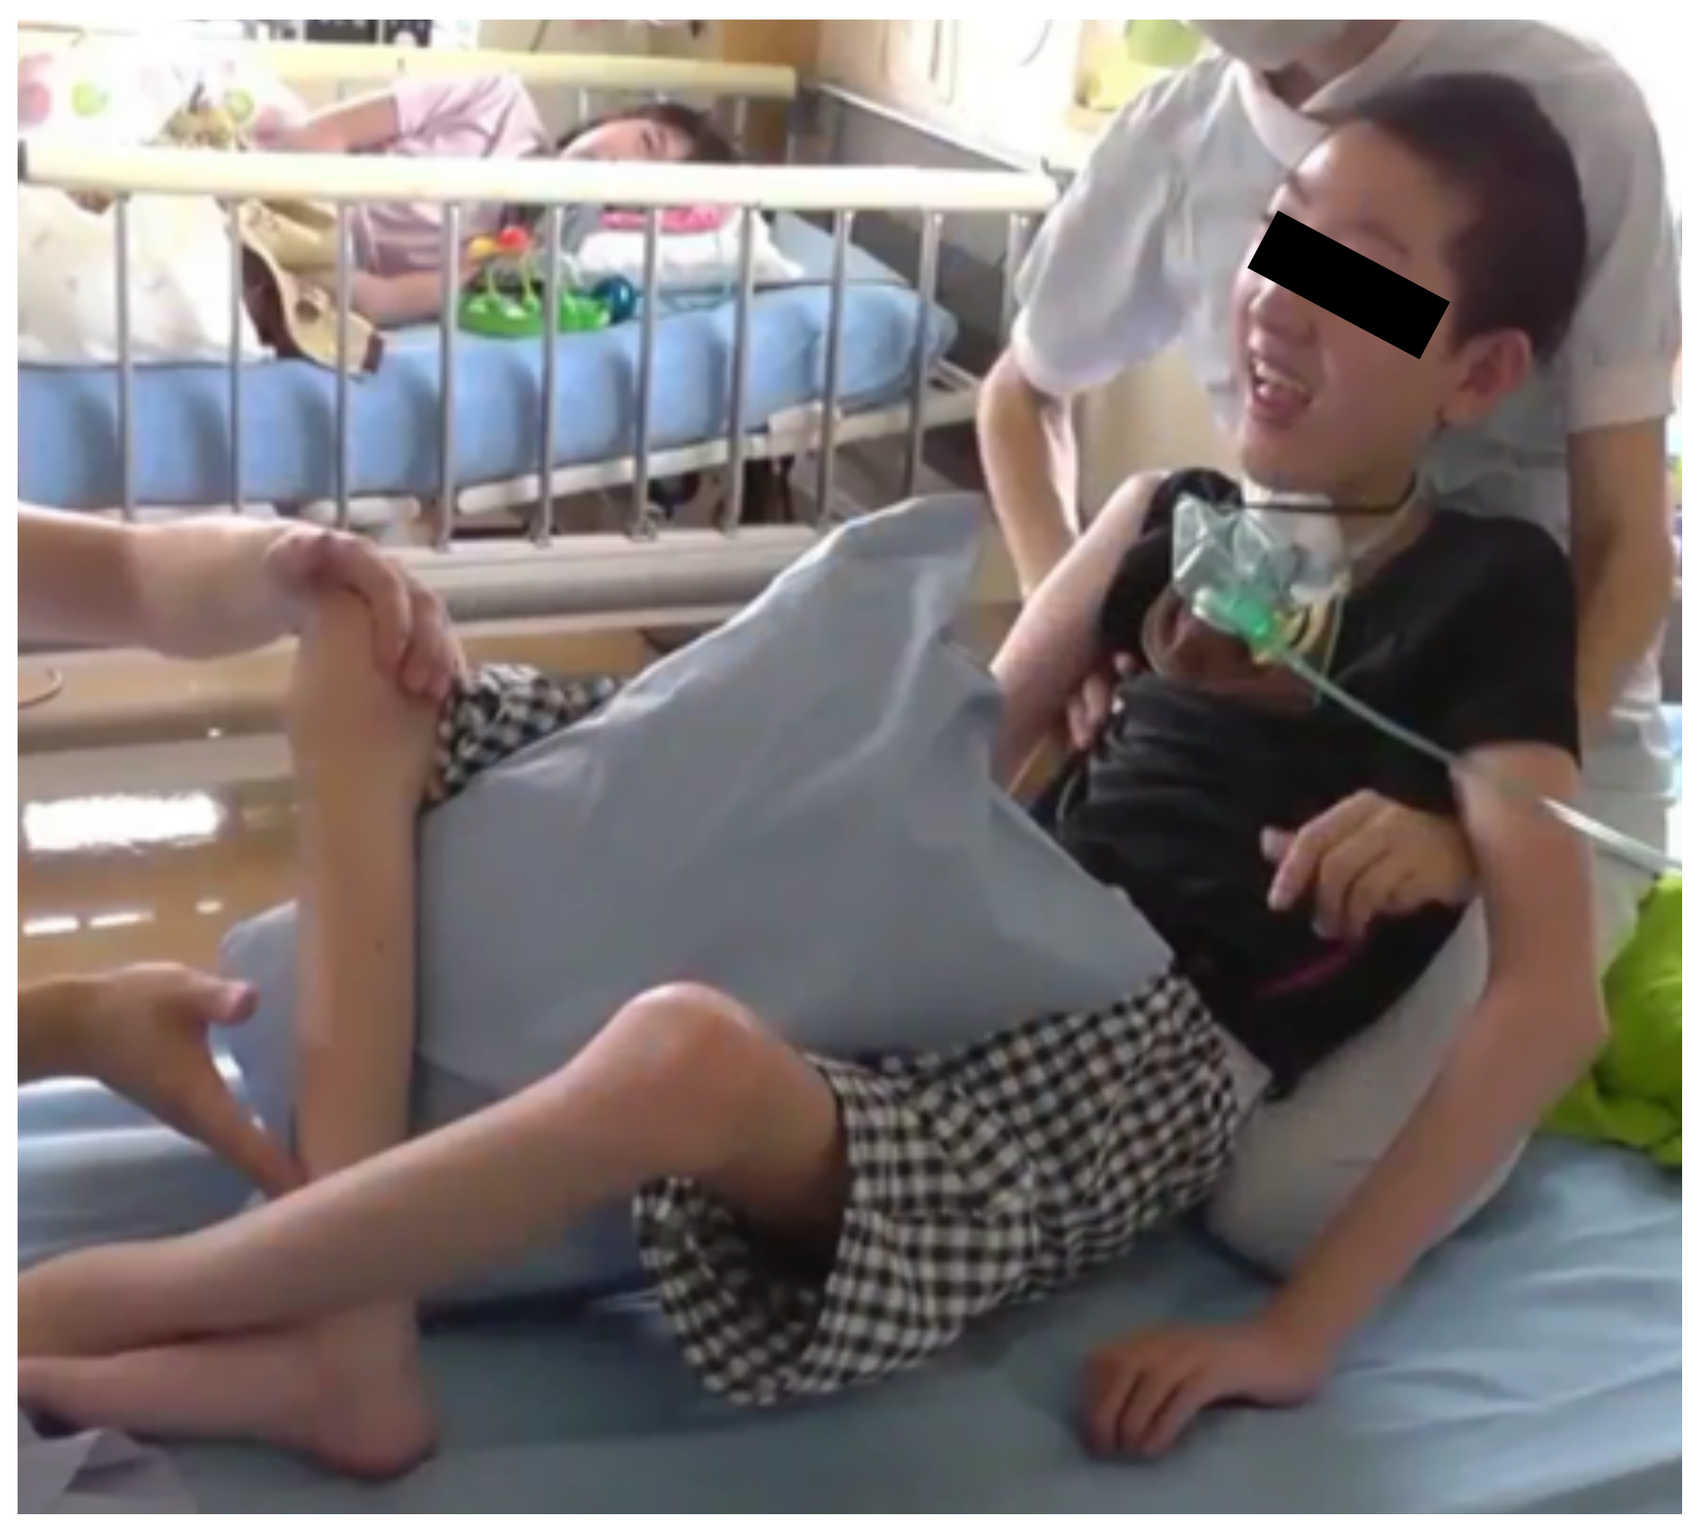

patient 2

Images courtesy of Dr. Muramatsu, Department of Pediatrics, Jichi Medical University, Japan

AADC is an enzyme required for biosynthesis of dopamine and serotonin. AADC deficiency has a wide range of symptoms.

AADC is an enzyme required for biosynthesis of dopamine and serotonin[2,3,6,7]

- In the autosomal recessive disease AADC deficiency, mutations in the dopa decarboxylase (DDC) gene result in reduced AADC enzyme activity, leading to severe combined deficiency of the neurotransmitters dopamine, serotonin, norepinephrine, and epinephrine
- The result is an increase in L-dopa, 3-OMD, VLA, and 5-HTP, and a decrease in the neurotransmitter metabolites HVA and 5-HIAA

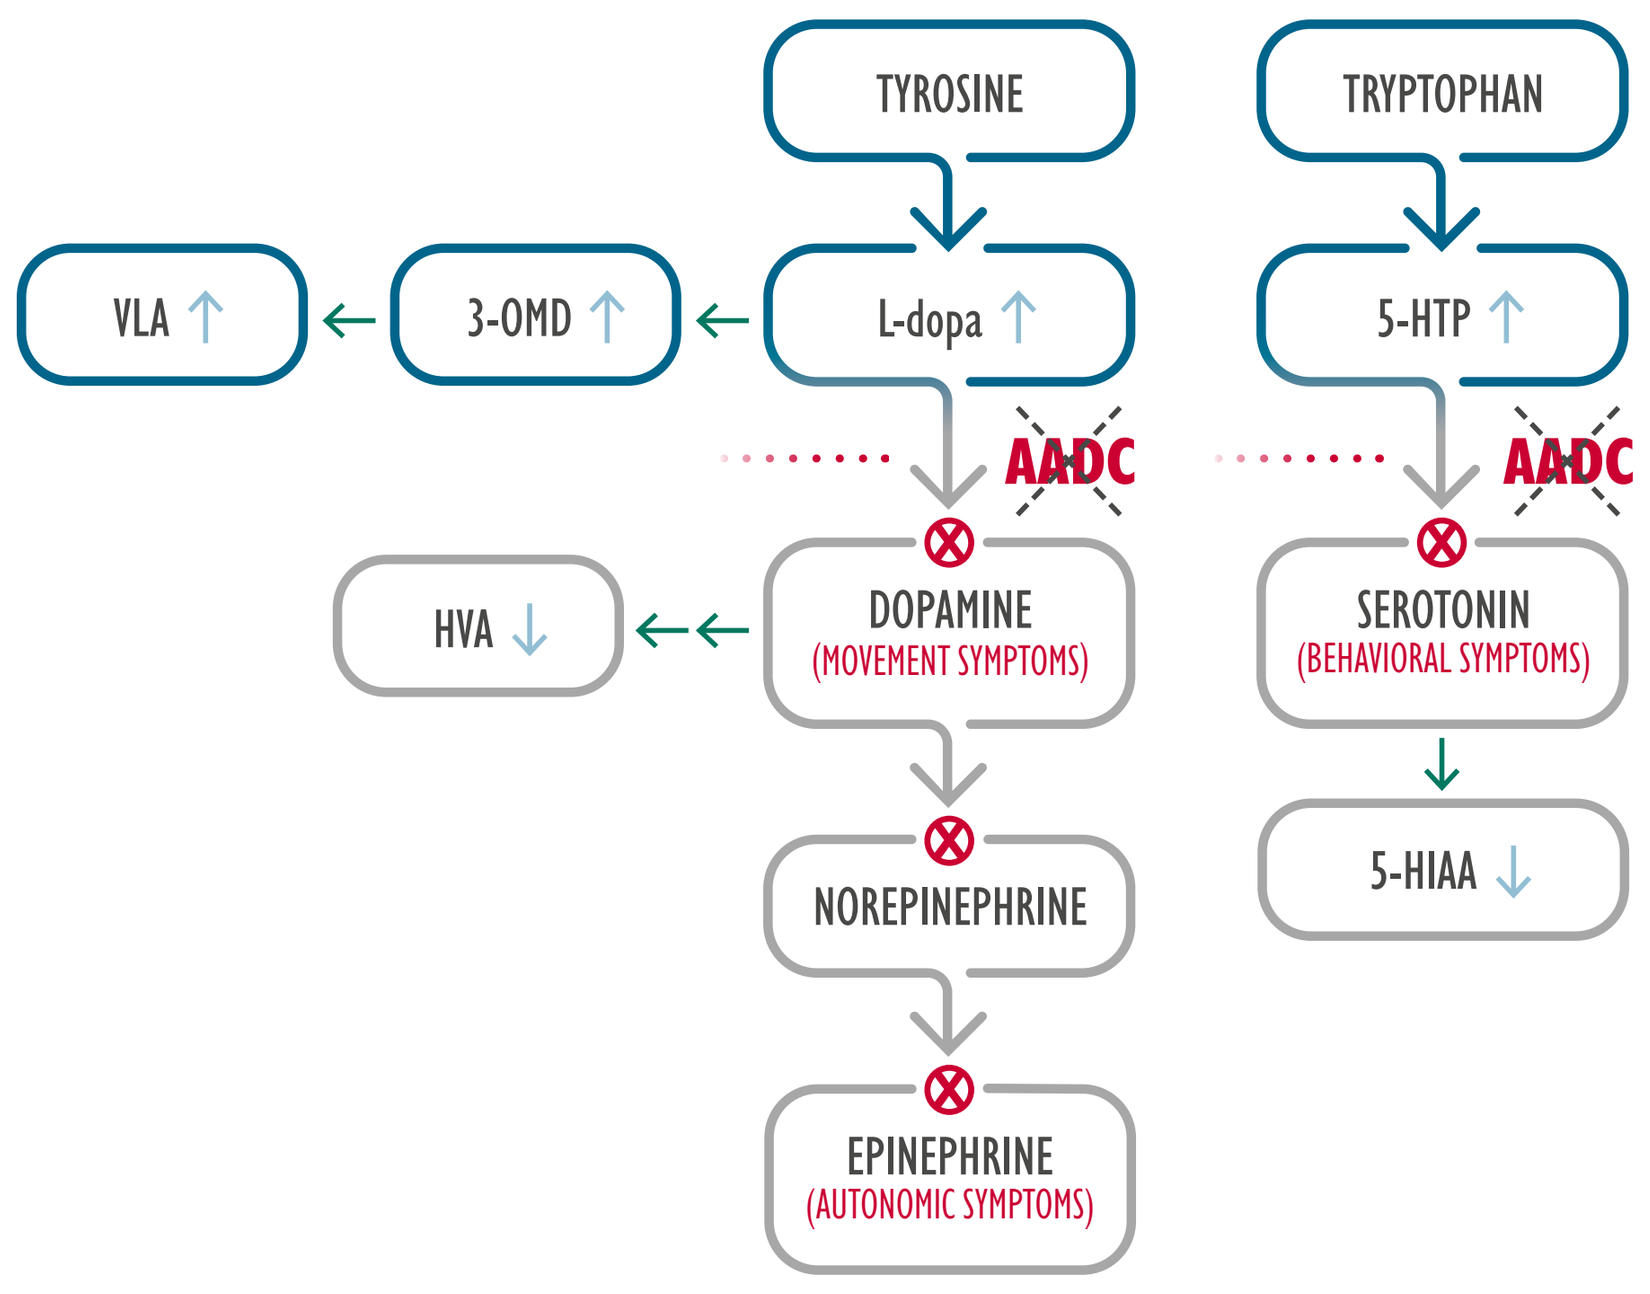
TYROSINE
TRYPTOPHAN
VLA ↑
3-OMD ↑
L-dopa ↑
5-HTP ↑
AADC
AADC
HVA ↓
DOPAMINE
(MOVEMENT SYMPTOMS)
SEROTONIN
(BEHAVIORAL SYMPTOMS)
NOREPINEPHRINE
5-HIAA ↓
EPINEPHRINE
(AUTONOMIC SYMPTOMS)

AADC deficiency can manifest with a broad spectrum of symptoms. The most common are[1-3]:

HYPOTONIA | DEVELOPMENTAL DELAY | MOVEMENT DISORDERS, ESPECIALLY OCULOGYRIC CRISIS

- In the published consensus guidelines:
 - Mean age of onset for patients with AADC deficiency was 2.7 months (n=68)[2]
 - Despite this young age of symptom onset, the mean age of diagnosis was 3.5 years (n=68)[2]

In a natural history study of 37 patients diagnosed with AADC deficiency, 97% failed to achieve motor milestones[1,5]

- This includes the ability to sit, stand, walk, run, and/or speak

Symptoms typically do not improve, leading to the need for lifelong care[1,3]

- Patients who live to adulthood suffer long-term cardiac and orthopedic complications and increased risk of infections[2]
- Body weight may be normal in the first few months, but growth and weight gain can slow after the first year[5]

AADC deficiency can affect patients of any gender, ethnic origin, or geographic region[2,5]

- AADC deficiency has been reported in individuals of Asian, Caucasian, Arabic, Iranian, and Jewish ethnic origin[2,a]

AADC deficiency is associated with specific signs and symptoms, but lack of correct diagnosis often delays treatment and proper management

Symptom presentation in patients with AADC deficiency (%)[3,a]

Hypotonia[2-5]
- Most commonly reported symptom

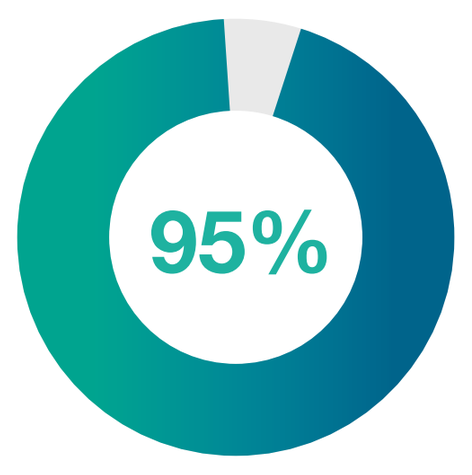
95%

Developmental delay[2-5]
- Impairments in head control, sitting, crawling, or standing
- Speech delays

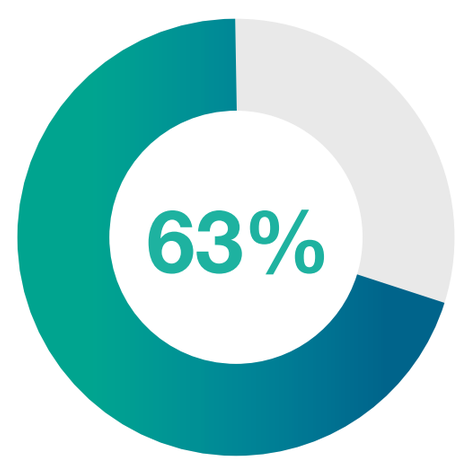
63%

Movement disorders

Oculogyric crisis[2,3,6,8]
- Episodes of sustained upward or lateral deviation of the eyes, rhythmic orofacial movements, backward and lateral flexions of the neck, tongue protrusion, and jaw spasms
- Can last a few seconds or persist for several hours, and occur several times per day or week
- May not be present in all cases

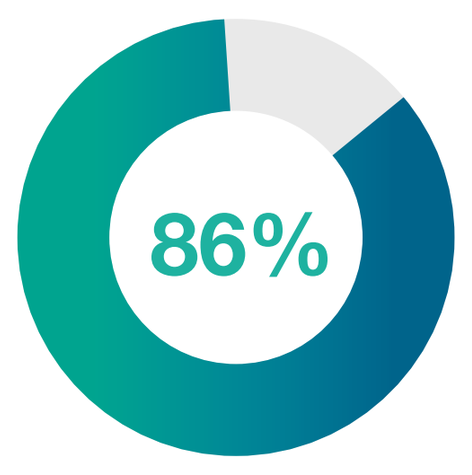
86%

Others[2,3]
- Dystonia (53%)
- Hypokinesia (32%)

Autonomic symptoms[2,3,6]
- Hyperhidrosis (65%)
- Ptosis (39%)
- Hypersalivation (41%)
- Nasal Congestion (31%)

[a]Percentages based on a clinical study of 78 patients diagnosed with AADC deficiency.[3]

Despite symptom onset during infancy, diagnosis is typically delayed[2,3]

3,5 — Mean age of diagnosis: 3.5 years[2]

Age range of diagnosis: 2 months to 23 years[2]

Many of the most common symptoms of AADC deficiency can also be attributed to a number of other conditions such as cerebral palsy and epilepsy, resulting in potential misdiagnosis.[2,3,9-11] However, the presence of the following should prompt investigation of AADC deficiency:

- Autonomic symptoms that are not typical of other conditions[2,8-11]
- Normal EEG and neuroimaging; one study showed that only a small proportion of patients with AADC Deficiency had an abnormal EEG, MRI, or CT[3]

References

1. Manegold C, Hoffmann GF, Degen I, et al. J Inherit Metab Dis. 2009;32(3):371-380.
2. Wassenberg T, Molero-Luis M, Jeltsch K, et al. Orphanet J Rare Dis. 2017;12(1):12. doi: 10.1186/s13023-016-0522-z.
3. Brun L, Ngu LH, Keng WT, et al. Neurology. 2010;75(1):64-71.
4. Kojima K et al, Gene therapy improves motor and mental function of aromatic L-amino acid decarboxylase deficiency, Brain 2019: 142;322-333.
5. Hwu WL, Chien YH, Lee NC, et al. JIMD Rep. 2018;40:1-6. doi: 10.1007/8904_2017_54.
6. Pons R, Ford B, Chiriboga CA, et al. Neurology. 2004;62(7):1058-1065.
7. Hwu WL, Lee NC, Chien YH, et al. Adv Pharmacol. 2013;68:273-284.
8. Lee WT. Epilepsy & Seizure. 2010;3(1):147-153. https://www.jstage.jst.go.jp/article /eands/3/1/3_1_147/_article/-char/en. Accessed December 18, 2018.
9. Krigger KW. Am Fam Physician. 2006;73(1):91-100.
10. Ng J, Papandreou A, Heales SJ, et al. Nat Rev Neurol. 2015; 11(10):567-584.
11. Kurian MA, Dale RC. Continuum (Minneap Minn). 2016;22(4 Movement Disorders):1159-1185.
12. Garcia-Cazorla A, Duarte S, Serrano M, et al. Mitochondrion. 2008;8(3):273-278.
13. Helman G, Pappa MB, Pearl PL. JIMD Rep. 2014;17:23-27.

Earlier identification of AADC deficiency can help improve the care and management of patients

The diagnostic pathway for suspected AADC deficiency[2,12]

Test for AADC Deficiency

if there is:

Hypotonia AND Delayed Motor Development AND MRI Inconsistent With Clinical Symptoms

Movement Disorders
› Oculogyric Crisis › Dystonia › Hypokinesia and/or Bradykinesia

Autonomic symptoms are often present
• Ptosis • Temperature Instability • Nasal Congestion

Following this diagnostic process can help diagnose AADC deficiency[2]

1. **Perform[2]**
 - CSF neurotransmitter metabolite panel
 - Plasma enzyme activity assay

 Other diagnostic tests that may be helpful[2]
 - Blood level measurement of 3-OMD
 - Urinary organic acid analysis

2. **Interpret[2]**
 - Reduced HVA, 5-HIAA, and MHPG; elevated 3-OMD, L-dopa, and 5-HTP; and normal pterins in CSF

 AND/OR

 - Low plasma AADC enzyme activity
 - Increased urinary VLA

3. **Confirm with genetic testing[2]**
 - Mutation(s) in the DDC gene

Blood plasma	CSF	Genetic testing
Decreased AADC enzyme activity	HIGH LEVELS OF 3-OMD, L-dopa, 5-HTP LOW LEVELS OF 5-HIAA, HVA Normal pterins	Variants in DDC gene

CSF=cerebrospinal fluid; MHPG=3-methoxy-4-hydroxyphenylglycol.

Consensus guidelines recommend confirming an AADC deficiency diagnosis with a genetic test[2]

CONCLUSIONS

AADC deficiency is a rare and devastating disorder of the central nervous system that typically presents early and may require lifelong care[1-3,13]. It can cause hypotonia, developmental delay, movement disorders and autonomic symptoms[2-7]

Earlier identification can help improve the care and management of patients with AADC deficiency[1-3]. However, AADC deficiency may be misdiagnosed or undiagnosed[2,8].

Hypotonia is the most commonly reported symptom of AADC deficiency. It is therefore necessary to consider testing for AADC deficiency in patients with unexplained hypotonia.

Poster 104 – Theme 3: Share, Care, Rare: Transforming care for rare diseases by 2030