Economic Burden of Care and Treatment Options for Patients With Rett Syndrome: Two Systematic Literature Reviews

POSTER 96

Omar Dabbous[1 *], Vanessa Taieb[2], Emna Abdennadher[3], Meryem Bouchemi[3], Justyna Chorąży[4], Katarzyna Borkowska[4], Veneta Georgieva[5], Bryan E. McGill[1], Thomas A. Macek[1], Benit Maru[1], Ramesh Arjunji[1]

[1]AveXis, Inc., Bannockburn, IL, United States; [2]Creativ-Ceutical, London, United Kingdom; [3]Creativ-Ceutical, Tunis, Tunisia; [4]Creativ-Ceutical, Kraków, Poland; [5]Creativ-Ceutical, Sofia, Bulgaria

OBJECTIVE

To identify clinical trials in Rett syndrome and assess the economic burden of care for patients with this condition

INTRODUCTION

- Rett syndrome is a rare, non-inherited, genetic, post-natal, neurological disorder that occurs almost exclusively in girls and leads to severe impairments in a child's development.[1] It is characterized by apparently normal development for the first 6 to 18 months of life, followed by the loss of acquired fine and gross motor skills and the ability to engage in social interaction, and the development of stereotypic hand movements[2]
- Rett syndrome affects approximately 1 in 9,000 girls under 12 years of age[3] and almost all cases of Rett syndrome are caused by a mutation in *MeCP2*, found on the X chromosome[4]
- Limited treatment options target symptoms and not the genetic cause of the disease, leaving a significant unmet need[5]

METHODS

- Two systematic literature reviews related to Rett syndrome were performed on 25 June 2018

1) ECONOMIC BURDEN

- Outcomes of interest were costs (direct and indirect medical costs), medical resource use (hospital admissions, length of stay, physician and specialist visit, medications) and non-medical resource use (lost productivity and homecare or caregiver's time)

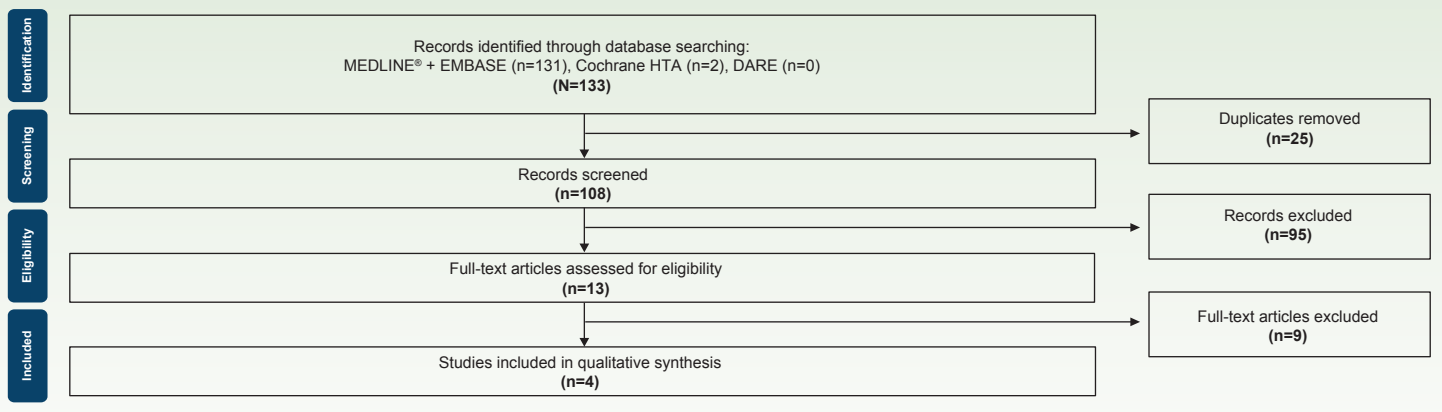
Identification
Screening
Eligibility
Included
Records identified through database searching:
MEDLINE® + EMBASE (n=131), Cochrane HTA (n=2), DARE (n=0)
(N=133)
Duplicates removed
(n=25)
Records screened
(n=108)
Records excluded
(n=95)
Full-text articles assessed for eligibility
(n=13)
Full-text articles excluded
(n=9)
Studies included in qualitative synthesis
(n=4)

2) TREATMENT OPTIONS

- Outcomes of interest were studies assessing the efficacy and safety of treatments for Rett syndrome

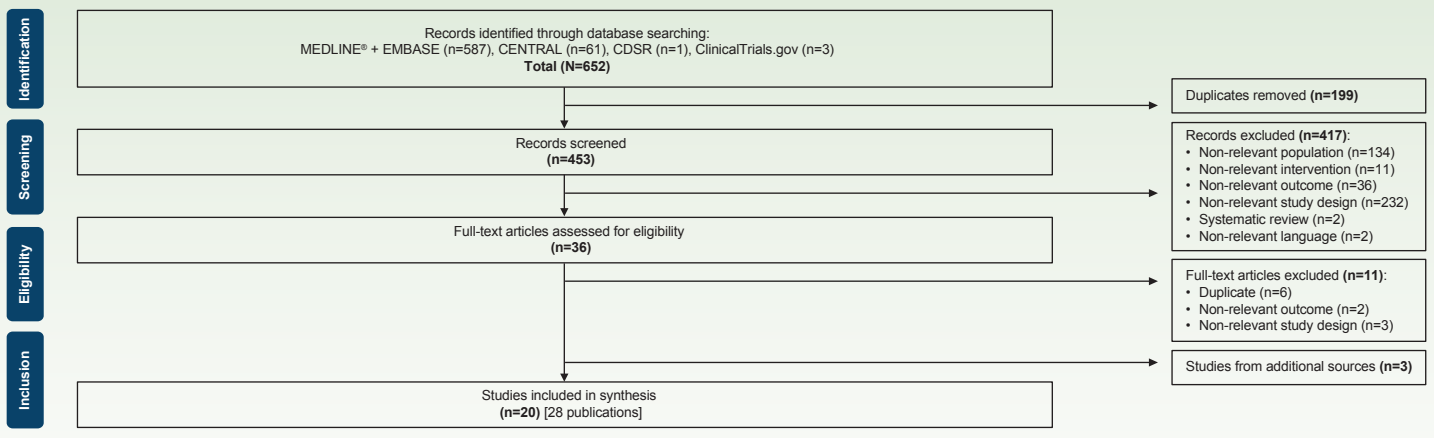
Identification
Screening
Eligibility
Inclusion
Records identified through database searching:
MEDLINE® + EMBASE (n=587), CENTRAL (n=61), CDSR (n=1), ClinicalTrials.gov (n=3)
Total (N=652)
Duplicates removed (n=199)
Records screened
(n=453)
Records excluded (n=417):
Non-relevant population (n=134)
Non-relevant intervention (n=11)
Non-relevant outcome (n=36)
Non-relevant study design (n=232)
Systematic review (n=2)
Non-relevant language (n=2)
Full-text articles assessed for eligibility
(n=36)
Full-text articles excluded (n=11):
Duplicate (n=6)
Non-relevant outcome (n=2)
Non-relevant study design (n=3)
Studies from additional sources (n=3)
Studies included in synthesis
(n=20) [28 publications]

RESULTS

ECONOMIC BURDEN

- Four studies were included in the qualitative synthesis out of 133 articles identified through database searching
- The 4 included studies were conducted in the US, UK, Canada, China, Australia, Switzerland, Ireland, and Italy and provided data on resource use and costs of Rett syndrome

STUDY DESIGN AND POPULATION CHARACTERISTICS

Study	Country	Population Overview	Study Design	Intervention(s)	Follow-up (Years)	Sample Size	Mean Age (Years)
Urion et al. (2016)[6]	US	Rett syndrome	Retrospective cohort	Care integration	1	–	–
Hammett et al. (2014)[7]	UK	Rett syndrome	Retrospective cohort	Surgical correction of scoliosis	2	11	12.6
MacKay et al. (2018)[8]	US, UK, Canada, Australia, China, Ireland, Switzerland	Rett syndrome	Cross-sectional	–	–	399	–
Labianca et al. (2013)[9]	Italy	Patients with Rett syndrome and neuromuscular scoliosis	Prospective cohort	Surgery (posterior spinal fusion and Luque or hybrid instrumentation)	Mean: 12.9 (2–24)	21	12.4

DIRECT MEDICAL RESOURCE USE RESULTS – LENGTH OF STAY

- Labianca et al., 2013[9], reported a mean length of stay of 12.3 days (range: 2–61) for patients with Rett syndrome and neuromuscular scoliosis treated by surgery
- According to Hammett et al., 2014[7]
 - A mean length of inpatient stay of 18.2 days (range: 8–42) was seen for 11 patients with Rett syndrome who underwent a surgical correction of scoliosis (posterior instrumented fusion of the curves)
 - Complications had considerable implications on the length of hospital stay; patients without complications were discharged on average 8.7 days postoperatively, whereas those with complications remained as inpatients for an average of 21.8 days
- MacKay et al., 2018[8], studied patients with confirmed *MeCP2* mutation and reported the shortest mean cumulative length of respiratory-related hospital stay was for patients younger than 7 years old and the greatest rate was in those 20 years or older over the 5-year exposure period

DIRECT COSTS – IMPACT OF CARE INTEGRATION

- Urion et al., 2016[6], reported a decrease in inpatient charges from 48% total spend to 41% over the first year of care integration, suggesting that this intervention reduces costs in a population with Rett syndrome

DIRECT MEDICAL RESOURCE USE – HOSPITAL ADMISSION

- Two studies (Urion et al. 2016[6] and MacKay et al. 2018[8]) reported hospital admissions for patients with Rett syndrome

Hospital admissions

Urion et al. (2016)[6]	MacKay et al. (2018)[8]
• Unanticipated hospital admissions decreased an average of 2.06 per person/year to 1.32 per person/year over the first year of care integration	• Hospital admission over a 5-year exposure period due to LRTI was reported for slightly more than one-fifth (21.4%) of individuals, with a total of 230 admissions • Mean rate of hospital admissions for LRTI was 0.12 (95% CI: 0.11–0.13) per person/year over 5 years • Enteral feeding increased the risk of hospital admissions for LRTI (adjusted relative risk[a] 1.79, 95% CI: 1.21–2.65) • Compared with independent walking, being unable to walk was associated with a 6-fold increased risk of respiratory-related hospital admissions (adjusted relative risk[b] 6.73, 95% CI: 3.42–13.25)

[a]Adjusted for age group and mutation type. [b]Adjusted for age group and impacts of breath holding and hyperventilation.

Mean cumulative length of stay (days per person/year) over the 5-year exposure period according to age (MacKay et al., 2018)[8]

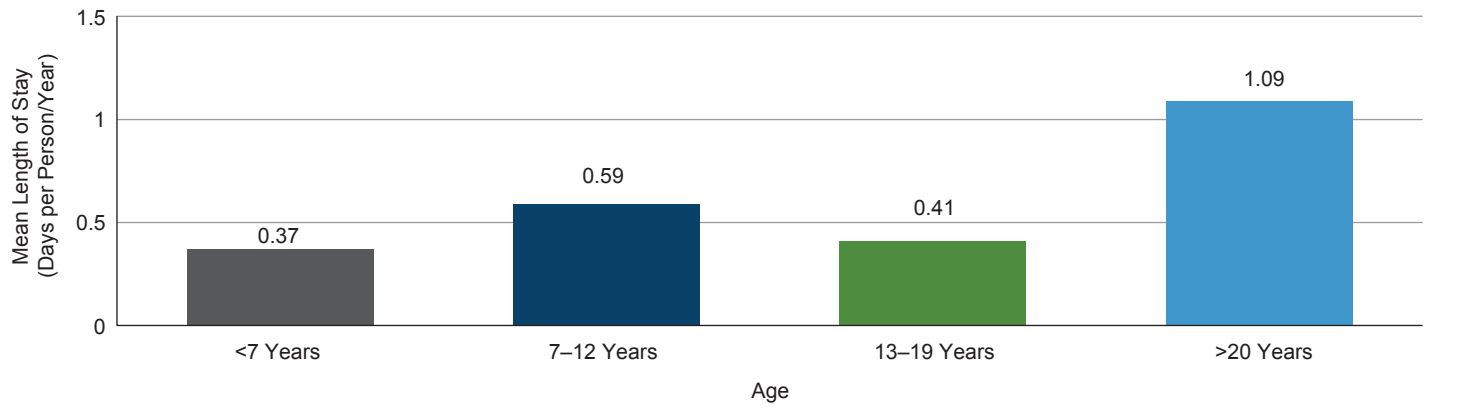
Mean Length of Stay (Days per Person/Year)
1.5
1
0.5
0
0.37
0.59
0.41
1.09
<7 Years
7–12 Years
13–19 Years
>20 Years
Age

Total cost of inpatient charges (M$) over 1 year of care integration (Urion et al., 2016)[6]

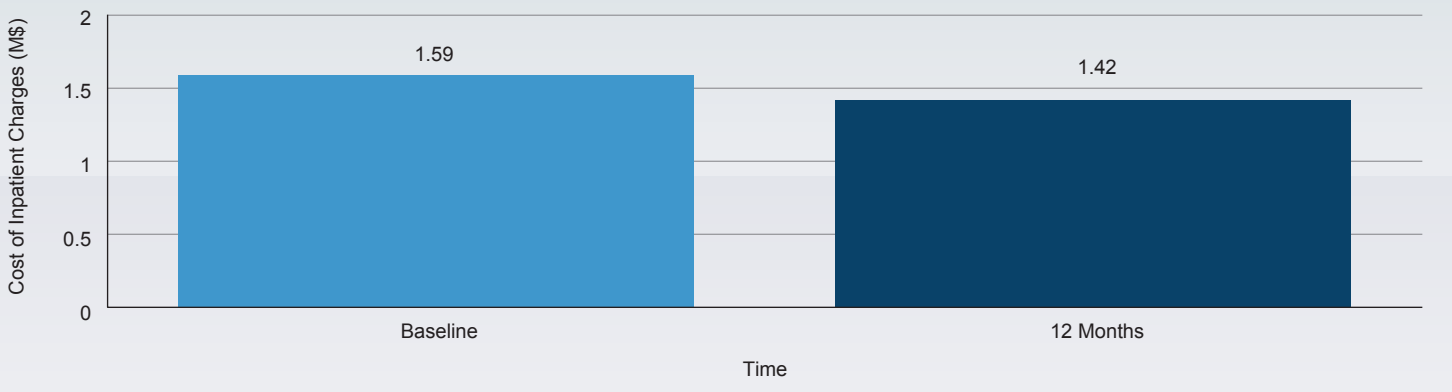
Cost of Inpatient Charges (M$)
2
1.5
1
0.5
0
1.59
1.42
Baseline
12 Months
Time

TREATMENT OPTIONS

- A total of 652 articles were identified, out of which 28 publications (20 studies) were included in the qualitative synthesis
- Out of 20 studies including from 8 to 82 identified patients, 19 focused on pharmacological treatments of Rett syndrome symptoms and 1 study examined the effects of environmental enrichment
- Overall, 5 (26%) were single-arm studies,[10–14] 15 (74%) were randomized controlled trials,[15–29] and 18 studies reported follow-up duration of 1 to 26 months

STATISTICALLY SIGNIFICANT RESULTS FROM CLINICAL STUDIES

Outcome Measure	Author, year	Intervention	Results (*P*-value)
RSBQ	Percy, et al. (2017)[18]	TFT 200 mg/kg vs PLC	**Clinical benefit was observed** for patients treated by **TFT** (0.042[T])
CGI	Percy, et al. (2017)[18]	TFT 200 mg/kg vs PLC	**Clinical benefit was observed** for patients treated by **TFT** (0.029[T])
VAS	Percy, et al. (2017)[18]	TFT 200 mg/kg vs PLC	**Clinical benefit was observed** for patients treated by **TFT** (0.025[T])
	O'Leary, et al. (2018)[22]	PLC-MCS vs MCS-PLC	**Worsening of symptoms** for patients treated by **PLC-MCS** (0.0211[T]; 0.0111[W])
ADAMS	O'Leary, et al. (2018)[22]	PLC-MCS vs MCS-PLC	**Worsening of symptoms** for patients treated by **PLC-MCS** (0.5535[T]; 0.0272[W])
EEG	O'Leary, et al. (2018)[22]	PLC-MCS vs MCS-PLC	**Worsening of symptoms** for patients treated by **PLC-MCS** (0.0208[T]; 0.0110[W])
	Gorbachevskaya, et al. (2001)[10]	CL vs UT	**Clinical benefit was observed** for patients treated with **CL**: lower value of LRP in alpha and beta bands (<0.001[T]; <0.01[T]), higher levels of LRP in the theta band (<0.001[T])
	Gorbachevskaya, et al. (2001)[10]	Before CL vs after CL	**Improvement of the brain functional stage** after treatment with **CL**: decrease of theta LRP in central and frontal regions (<0.05[T]; <0.01[T]), increase of beta activity LRP in the parietal region (<0.05[T]), restoration of occipital alpha rhythm (<0.05[T])
ECG	Guideri, et al. (2005)[25]	ALC at BS vs ALC after 6 mo	**Clinical benefit was observed** in patients treated with **ALC**: increase of total power (0.01[T]), VLF (0.01[T]), and LF (0.009[T])
	Guideri, et al. (2005)[25]	UT at BS vs UT after 6 mo	**Decrease in heart rate variability** was observed in **UT**: decrease of total power (0.04[T]) and LF (0.05[T]), and increase of QTcD (0.01[T])
Respiratory function	Percy, et al. (1994)[19]	NLT vs PLC	**Positive effect of NLT was observed:** higher awake min. O_2 saturation value (0.03[T]), less % time spent with disorganized breathing (0.02[T]), higher end tidal carbon dioxide value (0.02[T])
	Djukic, et al. (2016)[12]	Before glatiramer acetate 20 mg vs after	**Improvement of respiratory function:** decrease of breath hold index (0.004[T]; 0.03[W]) and breath hold time (0.007[T]; 0.004[W])
	Khwaja, et al. (2014)[13]	Pre MAD MCS vs post OLE	**Improvement of respiratory function:** improvement of apnea (0.012[T])
CSS	Maffei, et al. (2014)[26]	ω-3 PUFAs at BS vs ω-3 PUFAs after 6 mo	**Significant improvements were observed:** decrease in score for CSS (<0.005[A]), ambulation (0.02[A]), hand use (0.002[A]), motor (0.009[A]), non-verbal communication (0.002[A]), and respiratory dysfunction (<0.0001[A])

LIMITATIONS OF THE STUDIES

- In the economic burden analysis, studies by Urion et al., 2016[6], and Labianca et al., 2013[9], were published as conference abstracts. Findings should be interpreted cautiously as detailed information on these studies is lacking
- Almost half of the treatment options studies followed a crossover design and almost half had small sample sizes of 12 participants or fewer. Outcome data are limited, and evidence for PedsQL™, HAS, PEDI-m, and PEG use are lacking

CONCLUSIONS

- **Data on the costs of Rett syndrome management are sparse. Economic burden data were available only for direct medical resource use and direct medical costs; data on non-medical and indirect resource use and costs within the last 5 years were not identified**
- **There is an unmet need for safe and effective treatments for Rett syndrome**
- **Available treatment options address symptoms but do not improve disease progression**
- **Treatments that target the underlying cause of disease (e.g., gene-replacement therapy) could improve quality of life and overall prognosis for patients with Rett syndrome**
- **Further research is needed to better understand the impact of medical interventions for patients with Rett syndrome and their potential to reduce costs and/or increase utility**

ABBREVIATIONS: A, Analysis of variance; ADAMS, Anxiety, Depression, and Mood Scale; ALC, acetyl-L-carnitine; BS, baseline; CDSR, Cochrane Database of Systematic Reviews; CENTRAL, Cochrane Central Register of Controlled Trials; CGI, Clinical Global Impression; CI, confidence interval; CL, cerebrolysin; CSS, Clinical Severity Scale; DARE, Database of Abstracts of Reviews of Effects; ECG, electrocardiogram; EEG, electroencephalogram; EMBASE, Excerpta Medica Database; HAS, Hoffer Ambulation Scale; HTA, Health Technology Assessment; LF, low-frequency component (range: 0.04–0.15 Hz); LRP, logarithm of relative spectral power; LRTI, lower respiratory tract infection; MAD, multiple ascending dose; MCS, mecasermin; MCS-PLC, mecasermin for the first treatment period, placebo for the second; *MeCP2*, methyl-CpG binding protein 2; MEDLINE, Medical Literature Analysis and Retrieval System; mo, months; NLT, naltrexone; OLE, open-label extension; PEDI-m, Pediatric Evaluation of Disability Inventory; PedsQL™, Pediatric Quality of Life Inventory; PEG, percutaneous endoscopic gastronomy; PLC, placebo; PLC-MCS, placebo for the first treatment period, mecasermin for the second; PUFA, polyunsaturated fatty acids; QTcD, QTc dispersion (difference between the min. and max. heart rate–adjusted QT interval among the 12 ECG leads); RSBQ, Rett Syndrome Behavioural Questionnaire; T, student's t-test; TFT, trofinetide; UK, United Kingdom; US, United States; UT, untreated; VAS, visual analog scale; VLF, very low-frequency component (<0.04 Hz); W, Wilcoxon signed-rank test.

REFERENCES: 1. International Rett Syndrome Foundation. https://www.rettsyndrome.org/about-rett-syndrome. (Accessed: 30 March 2020). 2. Orphanet, https://www.orpha.net/consor/cgi-bin/OC_Exp.php?Lng=GB&Expert=778. (Accessed: 30 March 2020). 3. Anderson A, et al. *Orphanet J Rare Dis*. 2014;9:87. 4. United Kingdom National Health Service. https://www.nhs.uk/conditions/rett-syndrome. (Accessed: 30 March 2020). 5. AveXis. Press release, 17 June 2017. Available from: https://globenewswire.com (Accessed: 30 March 2020). 6. Urion D, et al. *Ann Neurol*. 2016;80(Suppl 20):S387. 7. Hammett T, et al. *Eur Spine J*. 2014;23(Suppl 1):S72– S75. 8. MacKay J, et al. *Dev Med Child Neurol*. 2018;60(9):951–957. 9. Labianca L, et al. *J Orthopaed Traumatol*. 2013;14(Suppl 1):S77. 10. Gorbachevskaya N, et al. *Brain Dev*. 2001;23:S90–S93. 11. Specchio N, et al. *Epilepsy Res*. 2010;88:112–117. 12. Djukic A, et al. *Pediatr Neurol*. 2016;61:51–57. 13. Khwaja OS, et al. *Proc Natl Acad Sci USA*. 2014;111:4596–4601. 14. Stenbom Y, et al. *Eur Child Adolesc Psychiatry*. 1998;7:49–52. 15. Downs J, et al. *Orphanet J Rare Dis*. 2018;13:3. 16. Ellaway C, et al. *J Child Neurol*. 1999;14:162–167. 17. Freilinger M, et al. *J Dev Behav Pediatr*. 2011;32:454–460. 18. Percy AK, et al. Presented at AACAP's 64th annual meeting. October 23–28, 2017. Washington, DC. 19. Percy AK, et al. *Ann Neurol*. 1994;35:464–470. 20. Glaze DG, et al. *J Child Neurol*. 2009;24:551–556. 21. MacArthur AJ, Budden SS. *Dev Med Child Neurol*. 1998; 40:186–192. 22. O'Leary HM, et al. *Ann Clin Transl Neurol*. 2018;5:323–332. 23. Smith-Hicks CL, et al. *Neurology*. 2017;89:1684–1690. 24. Glaze DG, et al. *Pediatr Neurol*. 2017:76:37–46. 25. Guideri F, et al. *Pediatr Cardiol*. 2005;26:574–577. 26. Maffei S, et al. *Mediators Inflamm*. 2014;983178. 27. Hagebeuk EE, et al. *J Child Neurol*. 2011;26:718–723. 28. Mancini J, et al. *Ann Clin Transl Neurol*. 2018;5:118–127. 29. Nielsen JB, et al. *Brain Dev*. 1990;12:143–147.

ACKNOWLEDGMENTS: The authors wish to thank the investigators and site acoordinators, and most importantly, the patients, families, and caregivers for their participation. Medical writing assistance was provided by Ashfield Healthcare Communications, part of UDG Healthcare plc, and was funded by AveXis, Inc, a Novartis company.

DISCLOSURES: This study was sponsored by AveXis, Inc., a Novartis company. OD, BEM, TAM, and RA are employees of AveXis, Inc., a Novartis company, and may hold Novartis stock or other equities. VT, EA, MB, JC, KB, and VG are employees of Creativ-Ceutical, a consultancy company working for various pharmaceutical industries, including AveXis, Inc. BM is an employee of SSI Strategy, who are contracted to support AveXis, Inc.

10th European Conference on Rare Diseases & Orphan Products 2020, Online, 14-16 May 2020