PO-028

NEXT GENERATION SEQUENCING (NGS) AN OPPORTUNITY FOR PATIENTS NON-DIAGNOSTIC IN LYSOSOMAL STORAGE DISEASE

Arévalo-Vargas I*[1,2,3], López de Frutos L[1,2], Serrano-Gonzalo I[1,2], Strunk M[4], Mozas P[3], Giraldo P[2]

(1) GIIS-012. Instituto de Investigación Sanitaria Aragón (IIS Aragón), Unidad de Investigación Traslacional, Hospital Universitario Miguel Servet, Zaragoza (50009), Spain; (2) Fundación para el Estudio y la Terapéutica de la Enfermedad de Gaucher y Otras Lisosomales (FEETEG) Zaragoza (50009), Spain. (3) Universidad de Zaragoza, Departamento de Bioquímica, Biología Molecular y Celular, Zaragoza (50009), Spain. (4) Instituto Aragonés de Ciencias de la Salud, Zaragoza (50009), Spain.

811305@unizar.es

Introduction

Diagnostic procedures based on new technologies allow mass population studies to be carried out with greater sensitivity and precision and with fast and acceptable costs (3). Lysosomal storage diseases (LSD) are a heterogeneous group (around 70) of rare, multisystem diseases, characterized by substrate accumulation due to an enzymatic deficiency in the majority of cases. Until now, the enzymatic analysis was the gold standard LSD screening programs (2). However, difficulty to identify patients can appear and therefore causing an uncompleted diagnosis (5). The introduction of new sequencing techniques permits the simultaneous study of different gene variants and enables more precise identification (4). In the last decades, the use of Next-Generation Sequencing (NGS) has allowed the sequencing of massive groups of genes in a particular experiment and has allowed the discovery of new genes associated with different genetic diseases.

Objectives

- ✓ Design a panel based to in the causal gene variants of LSD focused in the disorders with an approved treatment or with new therapies in clinical trials.
- ✓ Validate panel NGS with samples previously analyzed in laboratory.

Methods

1. Disorders with an approved treatment or with new therapies in clinical trials

LSD SELECTION

- Review list of LSD from LDN.
- Use LSD classification from Orphanet.
- Evaluation criteria publication include key words with lysosomal disease, treatment, clinical trial.

GENES LSD

- Pathways affected ORPHA: 68366.
- Publication review about the genes associated with LSD:
 - https://rarediseases.org/
 - https://www.nlm.nih.gov/
 - https://www.ncbi.nlm.nih.gov/pubmed/
 - ClinicalTrials.gov

INCLUDED REGIONS

- 5 'and 3' UTR
- Exons
- Exon-intron boundaries
- Deep intron with damaging genetic variants
- Promoter

2. Sample preparation

>11 Dried Blood Spot (DBS) of patients that diagnosed in our laboratory.

>DNA extraction: PrepFilerTM Kit (1)

3. Analysis

DNA SAMPLE › **LIBRARY** Adaptors Ligation Fragmentation › **SAMPLE LIBRARY** Single o paired-end › **SEQUENCING** Ion Torrent Genome MachineTM › **READ** NGS RESULTS IonReporter

Results

66 LSD → 22 LSD with approved treatment clinical trial → 27 genes related with those LSD

PATIENT	SAMPLE STATUS	GENE	DNA NUCLEOTIDE CHANGE		REFERENCE SEQUENCES
			Allele 1	Allele 2	
1	CARRIER	*GLB1*	c.918C>A	~	NM_000404.3
2	AFFECTED	*SMPD1*	c.1829_1831delGCC	c.1829_1831delGCC	NM_000543.5
3			c.1458T>A	c.1458T>A	
4	AFFECTED ♂	*GLA*	c.1046G>A	~	NM_000169.3
5	CARRIER ♀		c.155G>A	~	
6	AFFECTED	*NPC1*	c.2567T>C	c.3182T>C	NM_000271.5
7			c.1757delA	c.2746_2748del	
8	AFFECTED	*LIPA*	c.894G>A	c.894G>A	NM_000235.4
9			c.111+5G>C	c.894G>A	
10	AFFECTED	*GBA*	c.1226A>G	c.1448T>C	NM_000157.4
11			c.1226A>G	c.42_65del	

Table 1: Patients detected for NGS.

NGS method detected all variants in 11 previously studied patients (Table 1).

- **VALIDATION**

Despite the pseudogenes existence, damaging variants detection was correct.

- **PSEUDOGEN INTERFERENCE**

Conclusions

- Our NGS panel detect properly damaging variants on included genes.
- Variants were properly detected even when pseudogenes existed.
- The design of an NGS panel covering the treatable LSD diseases is a good approach to improve its diagnosis.

What is next?

The next step is to analyze neonatal sample of DBS collected

Funding: This work has been carried out with aid for research SHIRE International IIR-ESP-000140 and FEETTEG

THE 10th EUROPEAN CONFERENCE ON RARE DISEASES & ORPHAN PRODUCTS ecrd Stockholm, 15-16 May 2020

Instituto de Investigación Sanitaria Aragón

FEETEG

Departamento de Bioquímica y Biología Molecular y Celular Universidad Zaragoza 1542

References:

1. Brevnov, M. et al,. (2009). Developmental Validation of the PrepFilerTM Forensic DNA Extraction Kit for Extraction of Genomic DNA from Biological Samples. J Forensic Sci. Vol. 54, No. 3. 599-607.
2. Kwon JM, Matern D, Kurtzberg J, Wrabetz L, Gelb MH, Wenger DA, Ficicioglu C, Waldman AT, Burton BK, Hopkins PV, Orsini JJ. Consensus guidelines for newborn screening, diagnosis and treatment of infantile Krabbe disease. Orphanet J Rare Dis. 2018;13(1):30.
3. Neveling, K., Feenstra, I., Gilissen, C., Hoefsloot, L. H., Kamsteeg, E. J., Mensenkamp, A. R., … Nelen, M. R. (2013). A Post-Hoc Comparison of the Utility of Sanger Sequencing and Exome Sequencing for the Diagnosis of Heterogeneous Diseases. *Human Mutation*, *34*(12), 1721–1726. https://doi.org/10.1002/humu.22450
4. Rodríguez-Santiago, B., & Armengol, L. (2012). Tecnologías de secuenciación de nueva generación en diagnóstico genético pre- y postnatal. *Diagnostico Prenatal*, *23*(2), 56–66. https://doi.org/10.1016/j.diapre.2012.02.001
5. Sawyer, S. L., Hartley, T., Dyment, D. A., Beaulieu, C. L., Schwartzentruber, J., Smith, A., … Boycott, K. M. (2016). Utility of whole-exome sequencing for those near the end of the diagnostic odyssey: Time to address gaps in care. *Clinical Genetics*, *89*(3), 275–284. https://doi.org/10.1111/cge.12654