Poster 1

Genomic Testing in the Emerging Era of Precision Medicine: Lessons Learned from Studies in Larotrectinib

Jo Ballot[1], Lauren Kaplanis[2], Suzanne Ezrre[3], Tatiana Norman-Brivet[4], Genevieve Kelly[5], Sandra Montez[6]

[1]St. Vincent's University Hospital, Dublin, Ireland; [2]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [3]Dana-Farber Cancer Institute, Boston, MA, USA; [4]Stanford Comprehensive Cancer Center, Stanford, CA, USA; [5]Seattle Children's Hospital Cancer and Blood Disorders Center, Seattle, WA, USA; [6]MD Anderson Cancer Center, Houston, TX, USA

INTRODUCTION

- Identifying the genetic driver of a tumour may allow use of targeted therapies, which typically have fewer side effects than chemotherapies.
- Genomic testing can identify oncogenic alterations, including gene mutations and chromosomal rearrangements, that can be targeted with specific therapies.
- Chromosomal rearrangements may lead to gene fusions, resulting in novel oncogenic fusion proteins.[1]
 - Selective inhibitors of oncogenic fusion proteins enable precision medicine-based treatment of gene fusion cancers.[1]
 - Molecular testing is essential to identify patients, with rare tumour types, who may benefit from such agents (**Figure 1**).[2,3]

Figure 1. Molecular testing for gene fusions[2,3]

Next-generation sequencing (NGS)

- Tests for different genomic alterations in parallel using DNA or RNA
- RNA is preferred for gene fusions

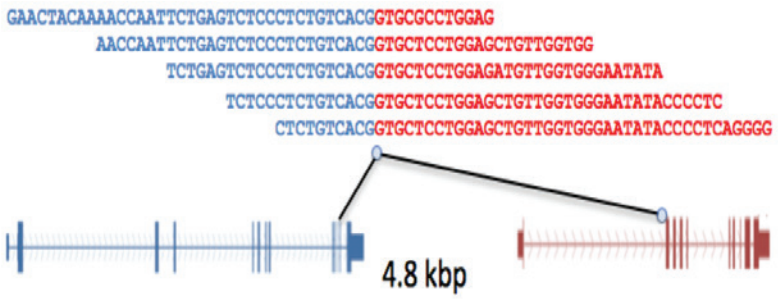
GAACTACAAAACCAATTCTGAGTCTCCCTCTGTCACGGTGCGCCTGGAG
AACCAATTCTGAGTCTCCCTCTGTCACGGTGCTCCTGGAGCTGTTGGTGG
TCTGAGTCTCCCTCTGTCACGGTGCTCCTGGAGATGTTGGTGGGAATATA
TCTCCCTCTGTCACGGTGCTCCTGGAGCTGTTGGTGGGAATATACCCCTC
CTCTGTCACGGTGCTCCTGGAGCTGTTGGTGGGAATATACCCCTCAGGGG
4.8 kbp

Reverse transcriptase polymerase chain reaction (RT-PCR)

- Detects known fusion transcripts in RNA
- Detects 5'/3' imbalance as a fusion signature, but cannot determine novel partners

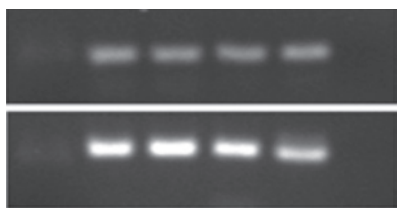

Fluorescence *in situ* hybridisation (FISH)

- Detects gene rearrangements in DNA that *may* generate a fusion transcript

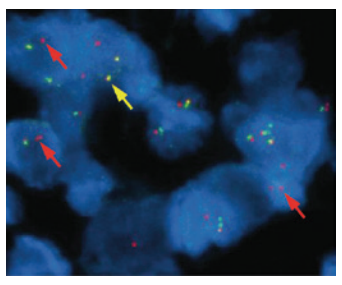

Immunohistochemistry (IHC)

- Detects protein expression, which *may* result from a fusion event

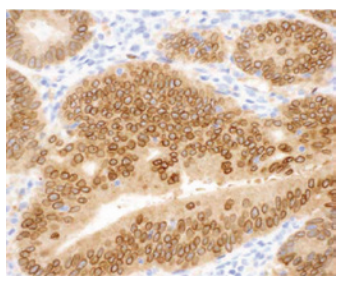

NTRK GENE FUSIONS

- Tropomyosin receptor kinase (TRK) fusions are oncogenic drivers of various tumours; they arise from rearrangements between neurotrophic tyrosine receptor kinase (*NTRK*) 1, 2, or 3 genes and an unrelated gene (**Figure 2**).[1]
- *NTRK* gene fusions identified in >20 paediatric and adult tumour types, and ~1% of all solid tumours.[1]
 - Rare in common cancers (e.g., lung, colon) but nearly pathognomonic in certain rare cancers (e.g., infantile fibrosarcoma, secretory carcinoma of the salivary gland, secretory breast carcinoma).

Figure 2. TRK fusions are rare but recurrent oncogenic drivers

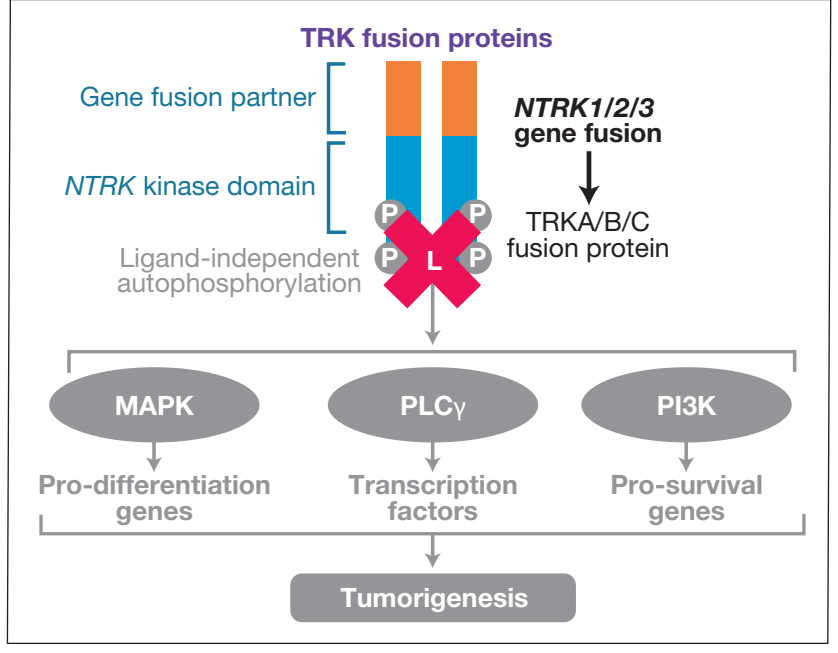
TRK fusion proteins
Gene fusion partner
NTRK kinase domain
NTRK1/2/3 gene fusion
TRKA/B/C fusion protein
Ligand-independent autophosphorylation
P
L
MAPK
PLCγ
PI3K
Pro-differentiation genes
Transcription factors
Pro-survival genes
Tumorigenesis

L, Larotrectinib.

- Larotrectinib is a highly selective, CNS-active TRK inhibitor approved in Brazil, Canada, the US and Europe to treat paediatric and adult patients with advanced solid tumours harbouring *NTRK* gene fusions.[1,4,5]
- In a pooled analysis of 159 patients across three phase I/II trials, objective response rate was 79% and median duration of response was 35.2 months.[6]
- Efficacy was seen regardless of tumour type (**Figure 3**) and age.[6]
- Larotrectinib was well tolerated. Most adverse events were mild (Grade 1) or moderate (Grade 2) in severity (**Table 1**).[6]

Figure 3. Change in tumour size on larotrectinib treatment[6]

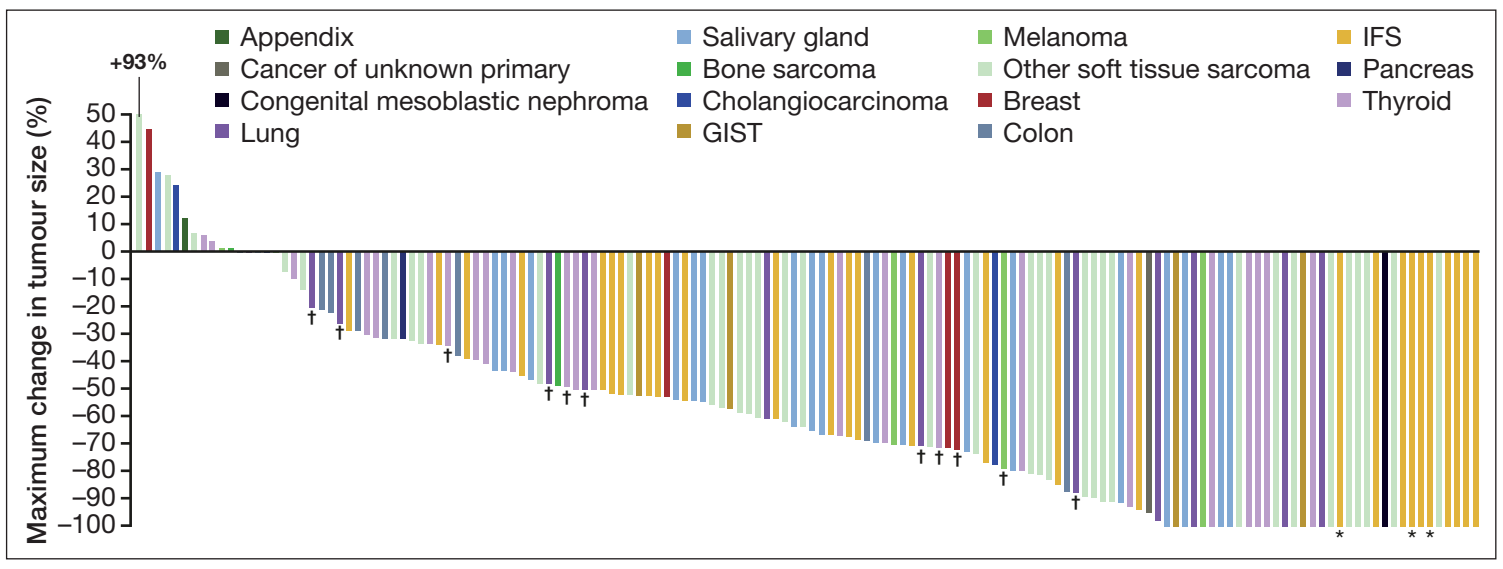
Appendix
Cancer of unknown primary
Congenital mesoblastic nephroma
Lung
Salivary gland
Bone sarcoma
Cholangiocarcinoma
GIST
Melanoma
Other soft tissue sarcoma
Breast
Colon
IFS
Pancreas
Thyroid
+93%
Maximum change in tumour size (%)
50
40
30
20
10
0
−10
−20
−30
−40
−50
−60
−70
−80
−90
−100

*Patients with a pathological complete response. †Patients with brain metastases.
GIST, gastrointestinal stromal tumour; IFS, infantile fibrosarcoma.

Table 1. Adverse events in the safety dataset (N=260)[6]

	Treatment-emergent AEs, (%)				Treatment-related AEs, (%)		
	Grade 1 or 2	Grade 3	Grade 4	Any grade	Grade 3	Grade 4	Any grade
Fatigue	30	2	0	33	<1	0	17
ALT increased	25	3	<1	28	3	<1	22
Cough	27	<1	0	28	0	0	1
Constipation	27	<1	0	27	0	0	11
Anaemia	17	10	0	27	2	0	10
AST increased	24	2	<1	27	<1	0	20
Dizziness	25	<1	0	25	<1	0	18
Nausea	24	<1	0	25	<1	0	13
Vomiting	24	<1	0	25	0	0	9
Diarrhoea	23	1	0	24	0	0	6
Pyrexia	19	<1	<1	20	0	0	2
Dyspnoea	13	2	0	16	0	0	<1
Myalgia	15	1	0	16	<1	0	8
Oedema peripheral	15	<1	0	16	0	0	6
Headache	15	<1	0	15	<1	0	5
Neutrophil count decreased	7	5	<1	12	2	<1	7

Grade 1/2 → Grade 3 → Grade 4
Mild/moderate → Severe → Life threatening

Adverse events that occurred at any grade in ≥15% of patients, or at grade 3 or 4 in ≥5% of patients, regardless of attribution, are listed. AE, adverse event; ALT, alanine aminotransferase; AST, aspartate aminotransferase.

CONSIDERATIONS OF GENOMIC TESTING

Benefits and drawbacks of different testing methods[2,3]

	NGS	FISH	RT-PCR	Pan-TRK IHC
BENEFITS	• Possible to detect novel fusion partners, and fusions expressed in RNA • Ability to test multiple actionable targets in parallel • Relevance of NGS increases as number of actionable targets grows • Potential for high sensitivity and specificity	• Location of target within cell is visible • High sensitivity and specificity • Several fluorophores can be used at once to detect multiple targets in one sample	• High sensitivity and specificity • Low cost	• Low cost • Identifies subcellular location of protein • Decentralised; available in most laboratories • Established reimbursement codes • Turnaround time: 1–2 days
DRAWBACKS	• Turnaround time: ~1–3 weeks • Technically complex and costly • Requires highly centralised testing model and specialty infrastructure • Reimbursement currently restricted • Sensitivity and specificity vary widely	• Requires fluorescence microscopy • Separate tests required for each *NTRK* gene • Cannot show that functional protein has been generated	• Target sequences must be known; cannot detect novel fusion partners • Separate tests required for each *NTRK* gene	• Cannot differentiate between fusion and wild-type TRK • Scoring algorithms not standardised • Confirmatory testing required

- Genomic testing requires accredited laboratories using approved platforms. In larotrectinib clinical trials, *NTRK* gene fusions were identified by NGS in most patients. Patients' tumours were tested in Clinical Laboratory Improvement Amendments-certified laboratories, or equivalent for ex-US sites.[7]

Examples of companies providing commercial NGS services	**Archer**	www.archerdx.com
	Caris Life Science	www.carislifesciences.com
	Foundation Medicine, Inc.	www.foundationmedicine.com
	Illumina	www.illumina.com
	Paradigm Diagnostics	www.paradigmdx.com
	PathGroup	www.pathgroup.com
	Tempus	www.tempus.com
	Thermofisher	www.thermofisher.com

- Panel of mutations tested for vary between companies and depend on which panel is ordered. NGS costs may be covered by private insurance or Medicare, and financial assistance programs may also be available.[8,9]

SUMMARY

- Genomic testing is essential and highly recommended to identify patients who may benefit from treatment with targeted therapies.
- Selective inhibitors of oncogenic fusion proteins can enable precision medicine-based treatment of patients with gene fusion cancers.
- Larotrectinib is effective and well tolerated in patients with TRK fusion cancer, regardless of age or tumour type. Rapid and reliable molecular testing is required to identify patients for larotrectinib therapy.
- The identification of *NTRK* gene fusions as actionable drug targets, highlights the importance of *NTRK* gene fusion testing across solid tumours.

References

1. Amatu A et al. *Ann Oncol*. 2019;30:viii5–viii15.
2. Penault-Llorca F et al. *J Clin Pathol*. 2019;72:460–467.
3. Wong D et al. *Pathol Oncol Res*. 2019 [Epub ahead of print].
4. Bayer HealthCare Pharmaceuticals Inc. VITRAKVI Prescribing Information 2018.
5. Bayer AG. VITRAKVI SmPC 2019.
6. Hong DS et al. *Lancet Oncol*. 2020;21(4):531–540.
7. Drilon A et al. *N Engl J Med*. 2018;378:731–739.
8. Foundation Medicine, Inc. Accessed Jan 30, 2019.
9. National Human Genome Institute. Accessed Jan 30, 2019.

Acknowledgements

We thank the patients and their families, many of whom traveled long distances to participate in these studies. The studies were funded by Bayer HealthCare Pharmaceuticals, Inc. and Loxo Oncology, Inc., a wholly owned subsidiary of Eli Lily and Company. Medical writing and editorial assistance was provided by Cindy Cheung, MD, and Annabel Ola, MSc, both of Scion (London, UK), funded by Bayer HealthCare Pharmaceuticals.

Presented at the 10th European Conference on Rare Diseases & Orphan Products, 14–15 May 2020

Presenting author: Jo Ballot, jo.ballot@ccrt.ie